3032 A Phase 1 Study of the Oral Administration of Irinotecan in Combination With the Potent P-glycoprotein (P-gp) Inhibitor Encequidar (NTC02250157)

Antonio Jimeno[1], Mateusz Opyrchal[2], Jennifer R Diamond[1], Christos Fountzilas[2], Bradley Corr[1], Ildiko Bezi[3], Hui Wang[3], Rudolf Kwan[4], Jay Zhi[3], David L Cutler[4], Patrick M Boland[2]

[1]University of Colorado, Aurora, CO; [2]Roswell Park Comprehensive Cancer Center, Buffalo, NY; [3]Athenex Inc, Cranford, NJ; [4]Athenex Inc, Buffalo, NY

INTRODUCTION

- Irinotecan is a prodrug of the potent topoisomerase inhibitor SN-38.
- The potential advantages of oral irinotecan administration include prolonged blood levels, less toxicity, greater convenience, reduced cost, and possible increase in the conversion to SN-38.
- Adenosine triphosphate-binding cassette transporter P-glycoprotein (P-gp) actively transports substrates such as various chemotherapeutic agents from intestinal epithelial cells back to the gastrointestinal tract and prevents the systemic absorption and intracellular accumulation of various cytotoxic agents.
- Many chemotherapies are substrates for P-gp and have poor oral bioavailability due to active efflux from the intestinal epithelium.
- Pre-administration of encequidar, a highly specific and potent blocker of P-gp, allows for absorption of chemotherapeutic agents across the intestinal epithelium and into the systemic circulation.
- Encequidar selectively binds to and inhibits P-gp, leading to an increase in both oral bioavailability and therapeutic efficacy.[1]
 - Encequidar is not systemically absorbed.[1]
- In animals, oral administration of irinotecan and encequidar increased the bioavailability of irinotecan.[1]

Figure 1. Encequidar: A Novel, Highly Specific, and Potent P-pg Inhibitor

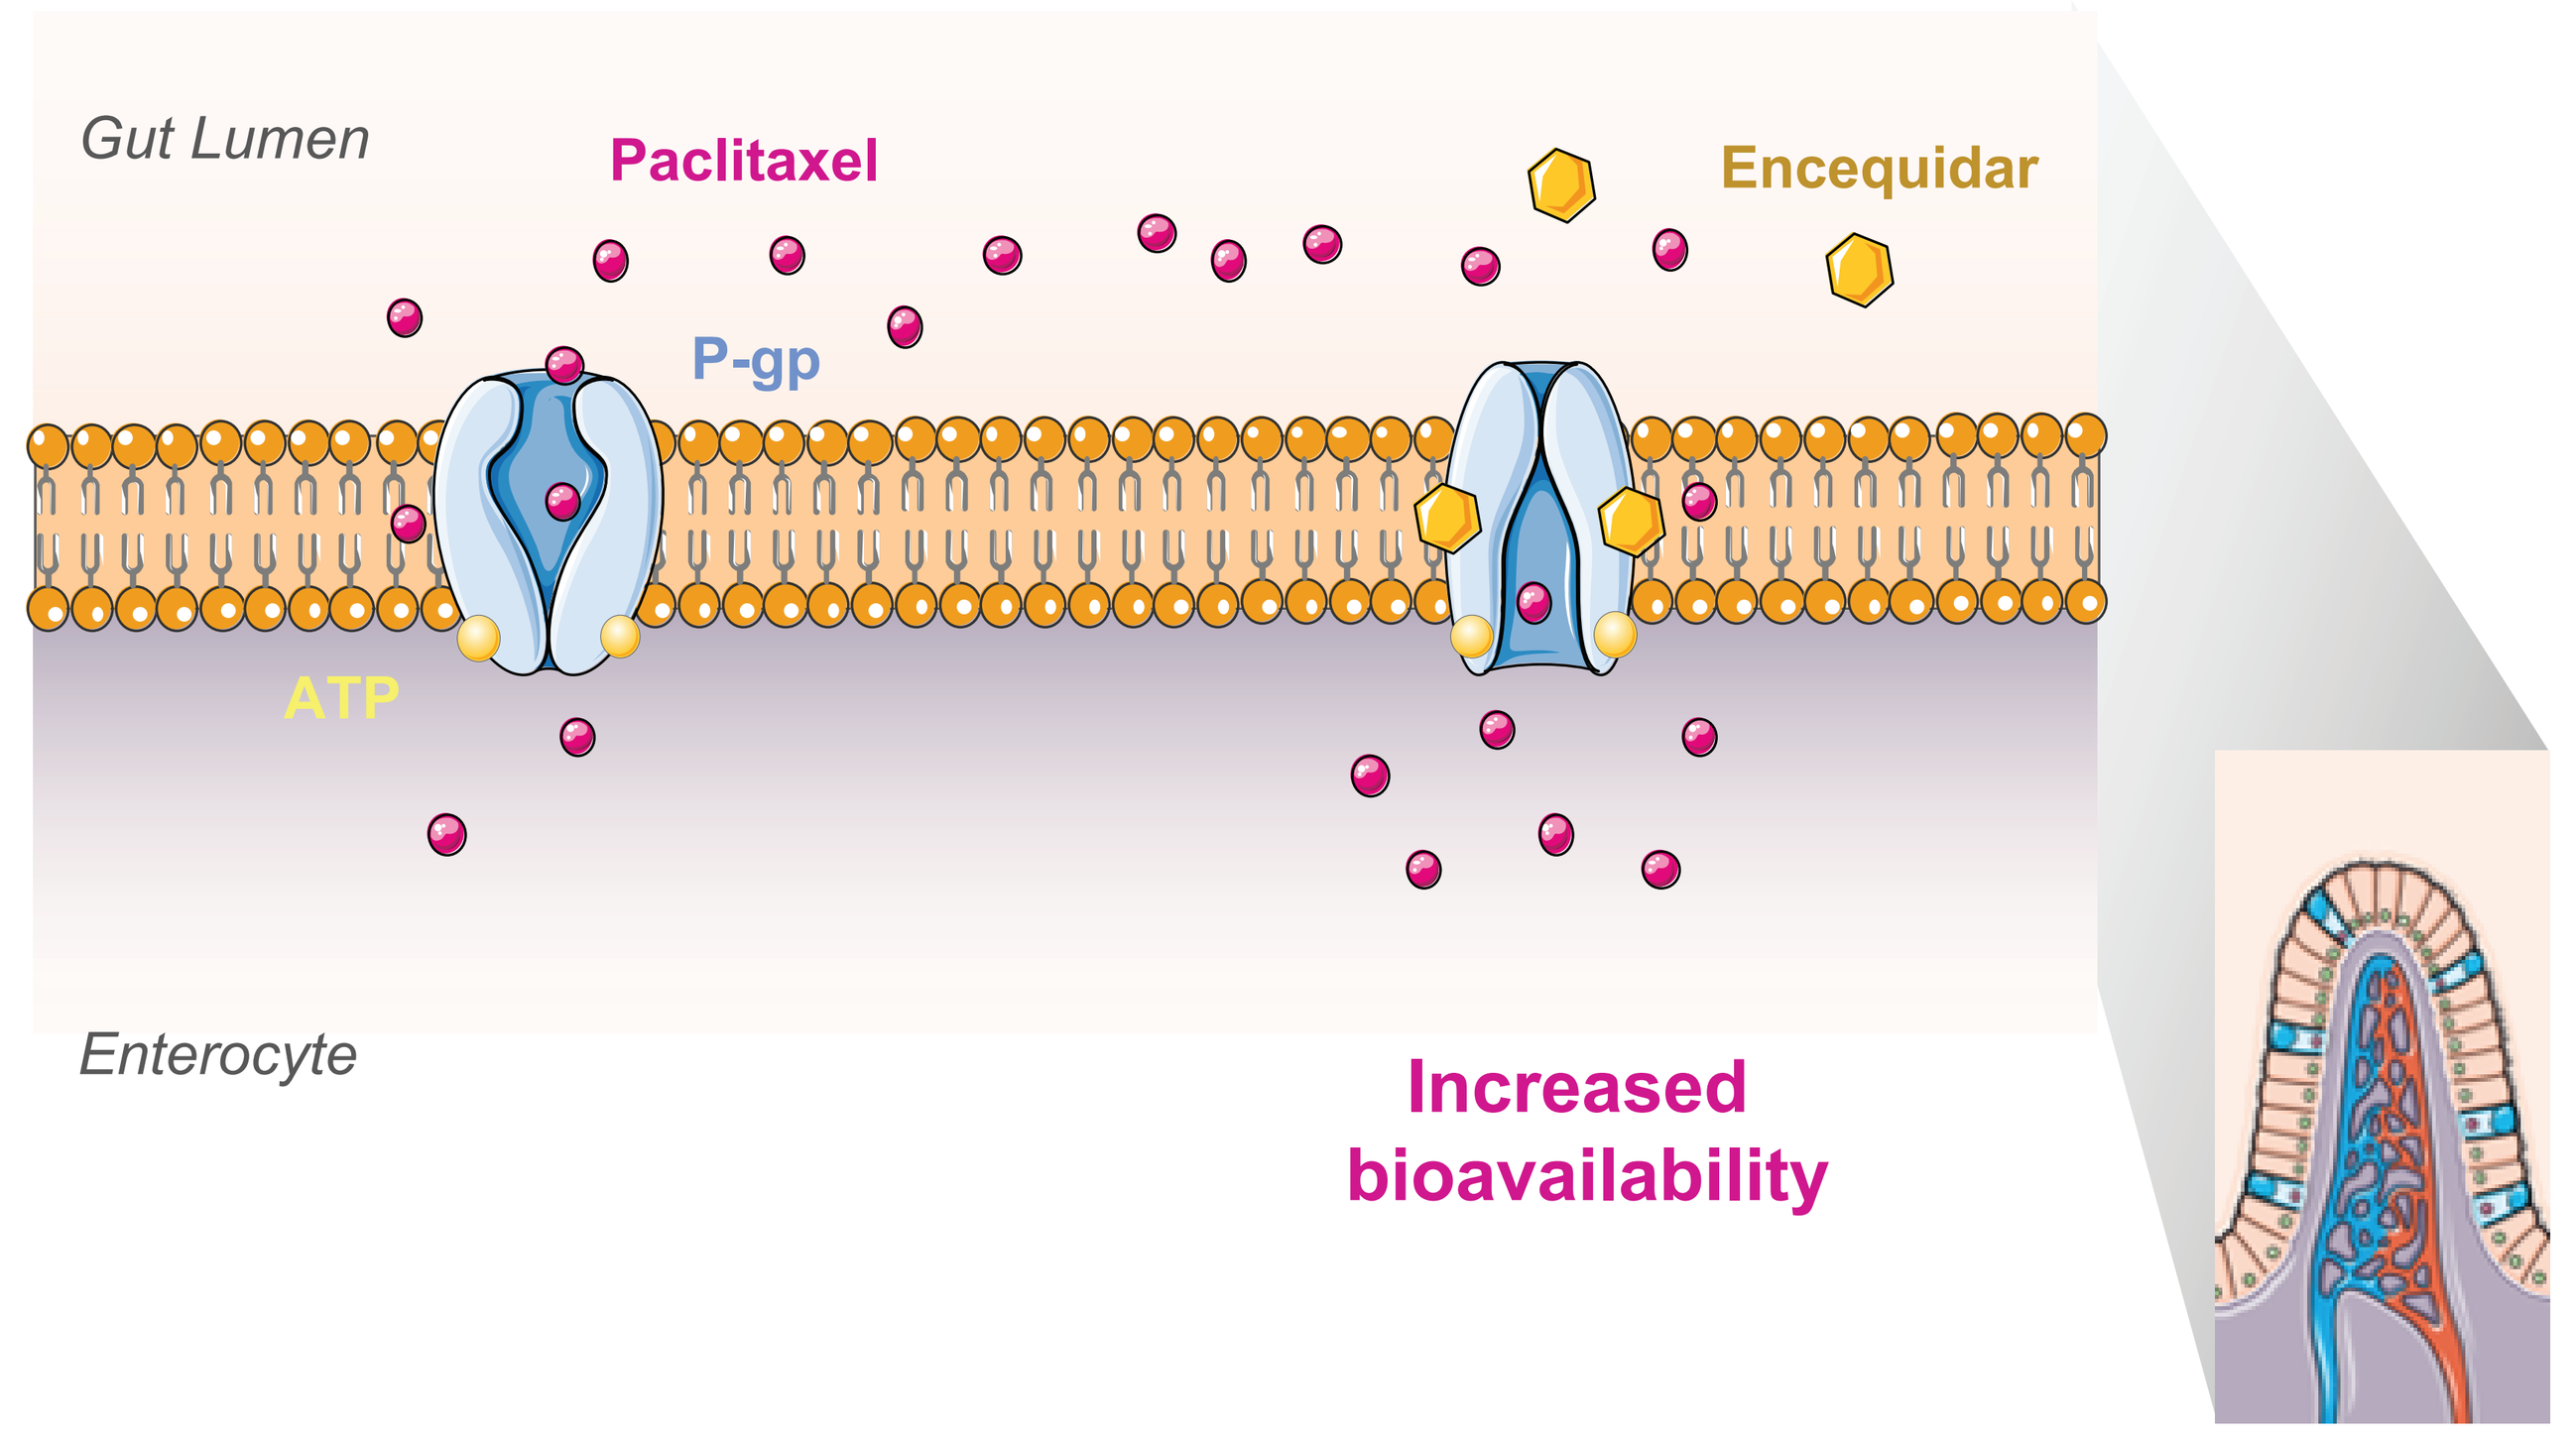
Gut Lumen
Paclitaxel
Encequidar
P-gp
ATP
Enterocyte
Increased bioavailability

Minimal / no systemic absorption	Localized P-gp inhibitory activity in GI tract	No significant systemic side effects	May reduce toxicities and improve clinical efficacy and tolerability vs IV therapies

GI, gastrointestinal; IV, intravenous; P-gp, P-glycoprotein.

OBJECTIVES

- The primary objective of this study was to determine the maximum tolerated dose and dose-limiting toxicity of orally administered irinotecan and encequidar on day 1 of a 21-day cycle.
- Secondary objectives were to determine the recommended phase 2 dose and the phamacokinetics (PK) of irinotecan and SN-38.

METHODS

- This was a phase 1 dose-escalation study enrolling cohorts of 3-6 patients with advanced malignancies.
- Patients had a hemoglobin level of ≥9 g/dL, an absolute neutrophil count of ≥1.5 × 10^9/L, a platelet count of ≥100 × 10^9/L, adequate hepatic and renal function, Eastern Cooperative Oncology Group Performance Status of 0-1, and were not homozygous for UGT1A1*28.
- Patients were administered, under fasting condition, encequidar 15 mg and oral irinotecan 20, 40, 80, 120, 160, 200, 240, 280, and 320 mg/m², initiated with a solution and then a tablet introduced at the 120 mg/m² dose level. n = 3 for all dose groups except 320 mg/m² dose (n = 6).
- PK samples for measurement of prodrug irinotecan and active SN-38/SN-38 glucuronide were collected up to 22 days (504 hours) post dose in cycle 1.

RESULTS

Baseline Demographics, Tumor Types, and Treatment Cycles Received in the Study

- Thirty male and female patients, mean age 60.9 years(range 33-78) were enrolled into this ongoing study.
- The most common cancers were ovarian (6), colorectal (4), breast (4), endometrial (3), and pancreatic (3).
- The median number of cycles administered was 3 (range 1-9).

Plasma Pharmacokinetics: Irinotecan

- Irinotecan plasma concentration-time profile follows biexponential decay at each dose level.
- Irinotecan exposure increases with dose, with the highest mean profile being achieved with the 320 mg/m² dose.

Figure 2.

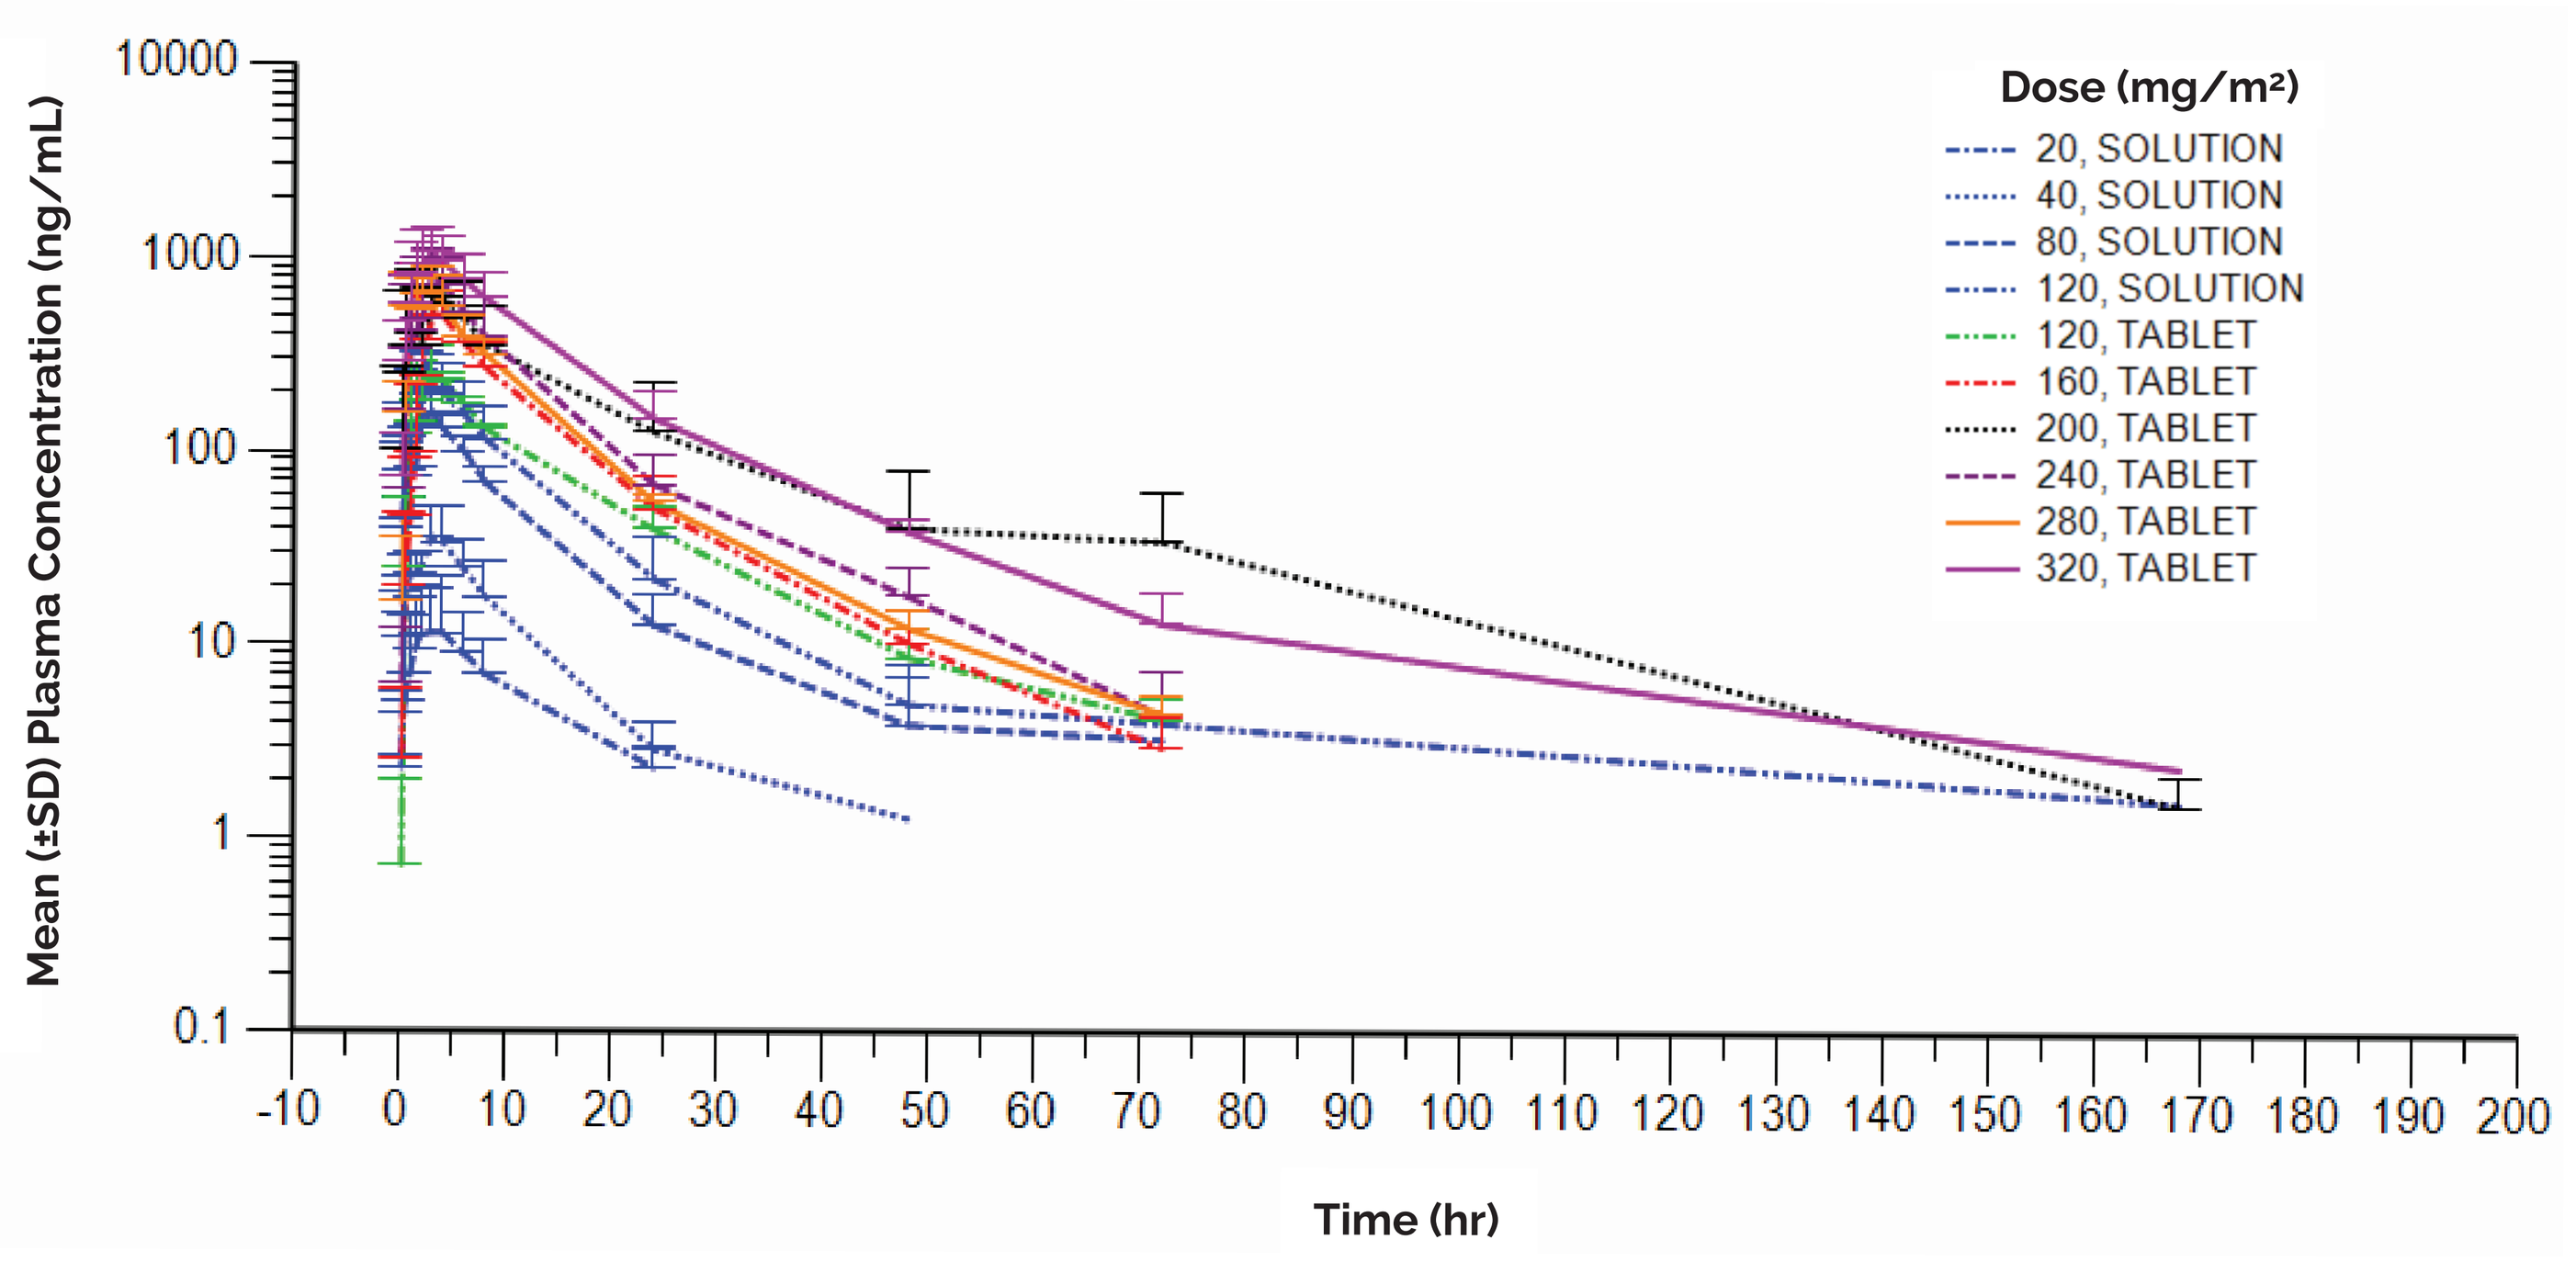
Dose (mg/m²)
20, SOLUTION
40, SOLUTION
80, SOLUTION
120, SOLUTION
120, TABLET
160, TABLET
200, TABLET
240, TABLET
280, TABLET
320, TABLET
Mean (±SD) Plasma Concentration (ng/mL)
Time (hr)

SD, standard deviation.

Plasma Pharmacokinetics: SN-38

- SN-38 plasma concentrations increase with dose.
- Terminal elimination half-life is ~20 hours.
- Enterohepatic recycling (EHR) of SN-38 is observed in the plasma concentration profile.
- Mean SN-38 profiles overlap at the highest 3 doses.

Figure 3.

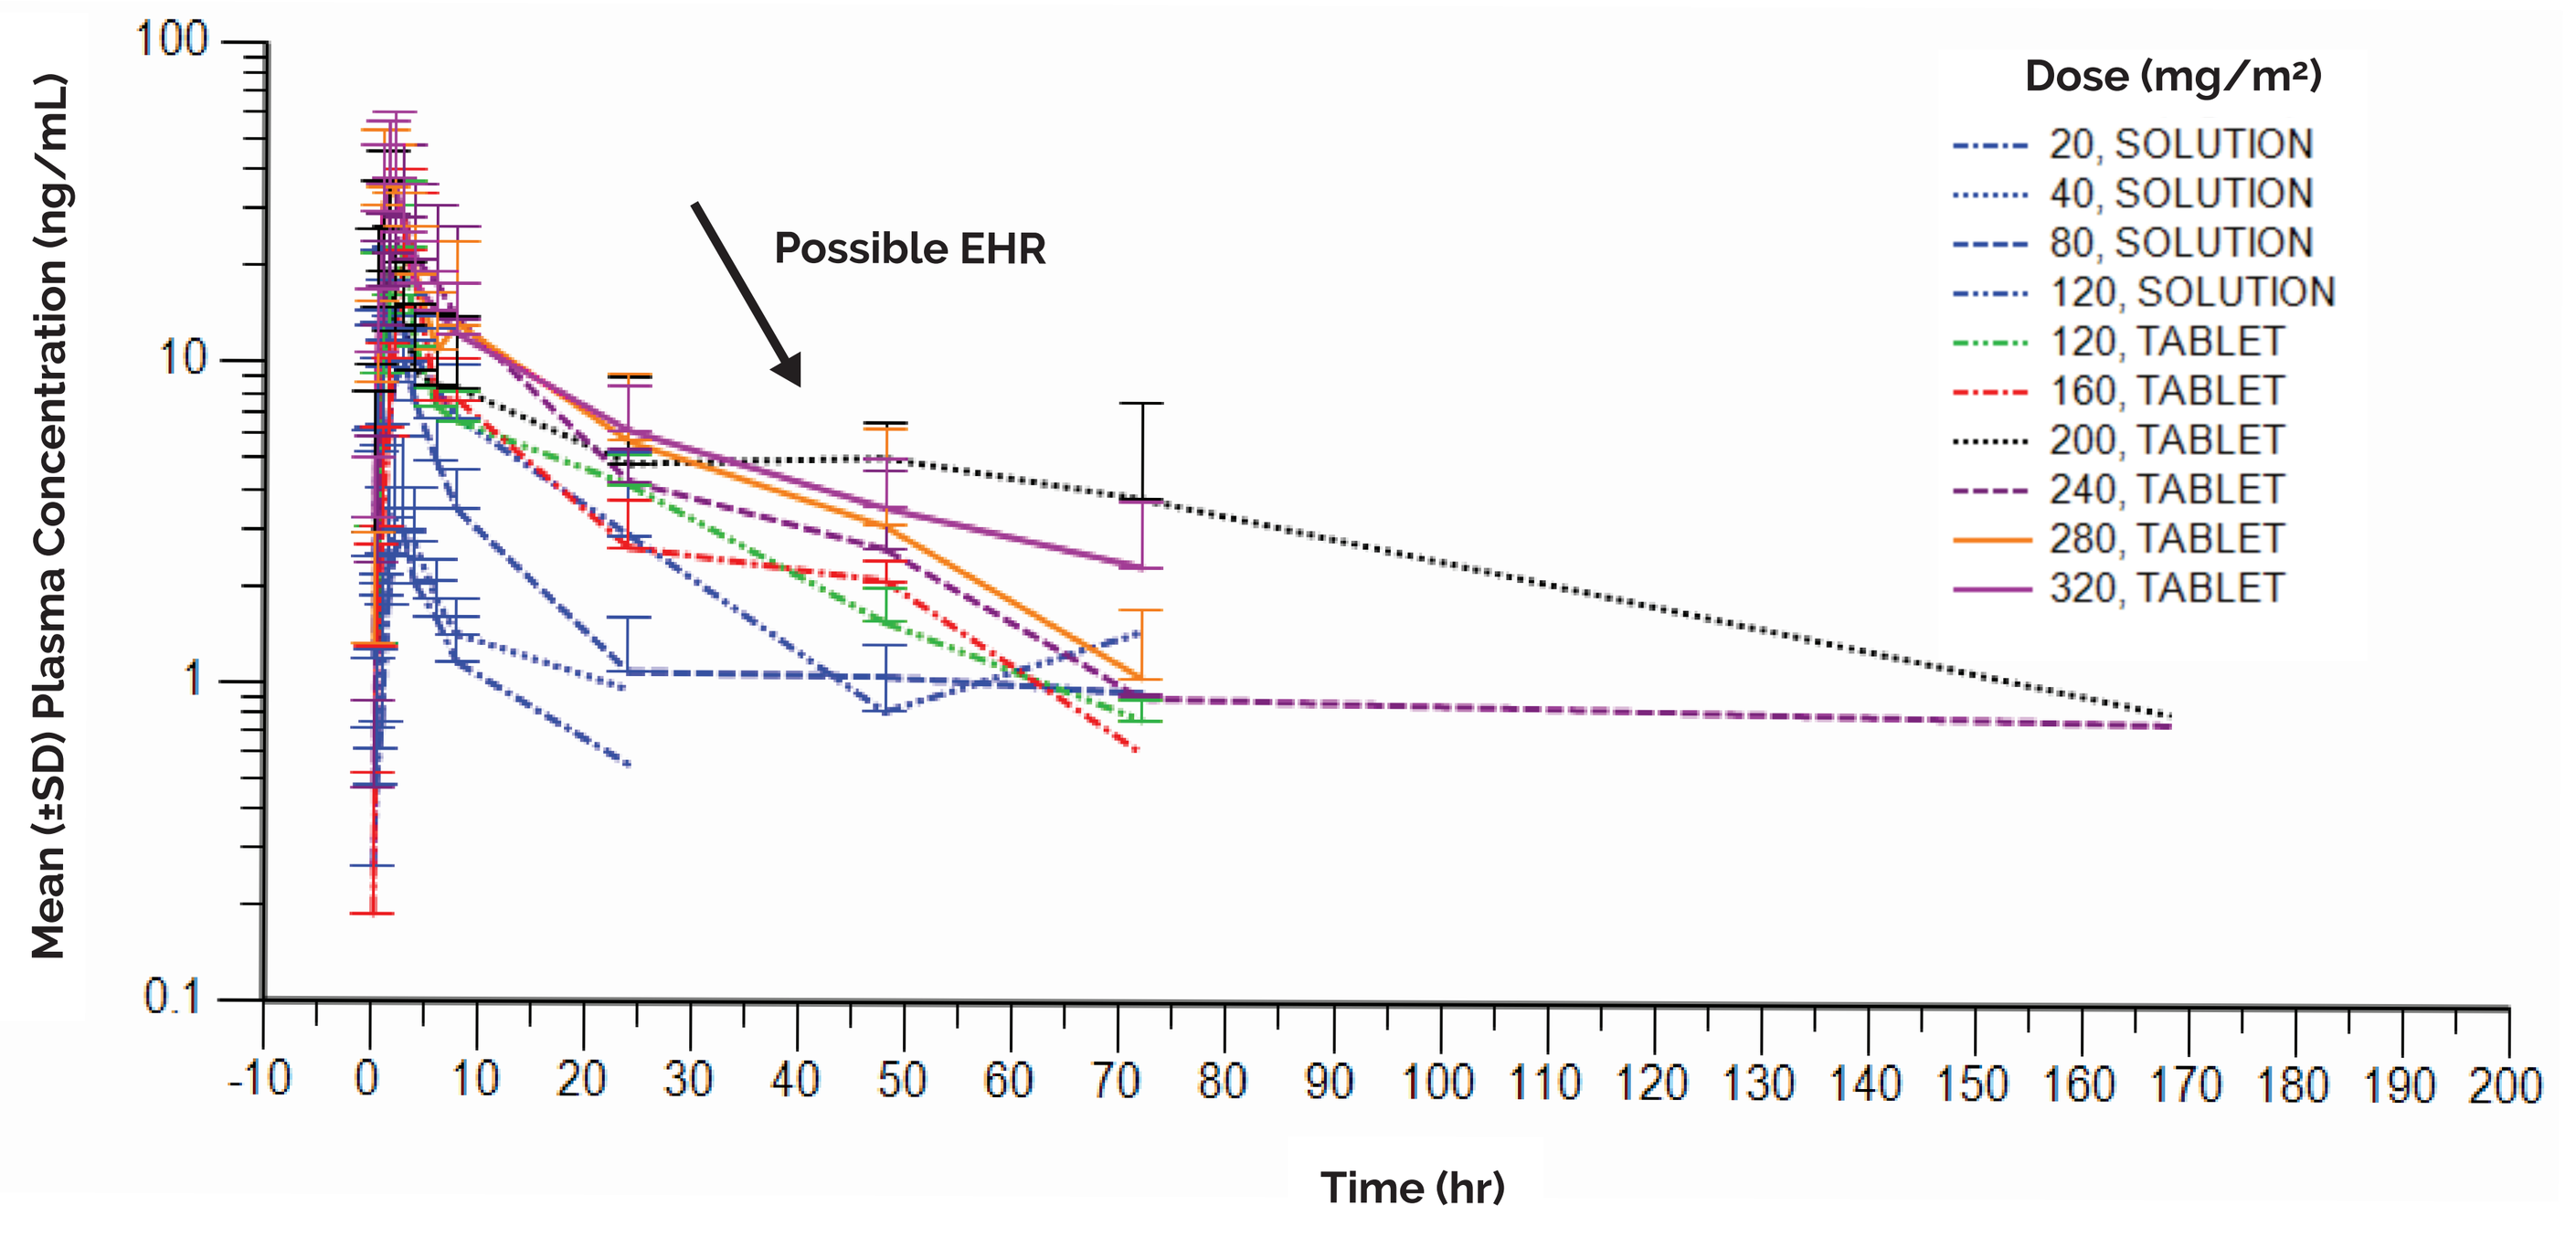
Possible EHR
Dose (mg/m²)
20, SOLUTION
40, SOLUTION
80, SOLUTION
120, SOLUTION
120, TABLET
160, TABLET
200, TABLET
240, TABLET
280, TABLET
320, TABLET
Mean (±SD) Plasma Concentration (ng/mL)
Time (hr)

EHR, enterohepatic recycling; SD, standard deviation.

Plasma Pharmacokinetics: C_{max} Comparison

- SN-38 C_{max} is variable across all dose levels for tablet formulation.
- SN-38 C_{max} is in range of that observed with intravenous Irinotecan.

Figure 4.

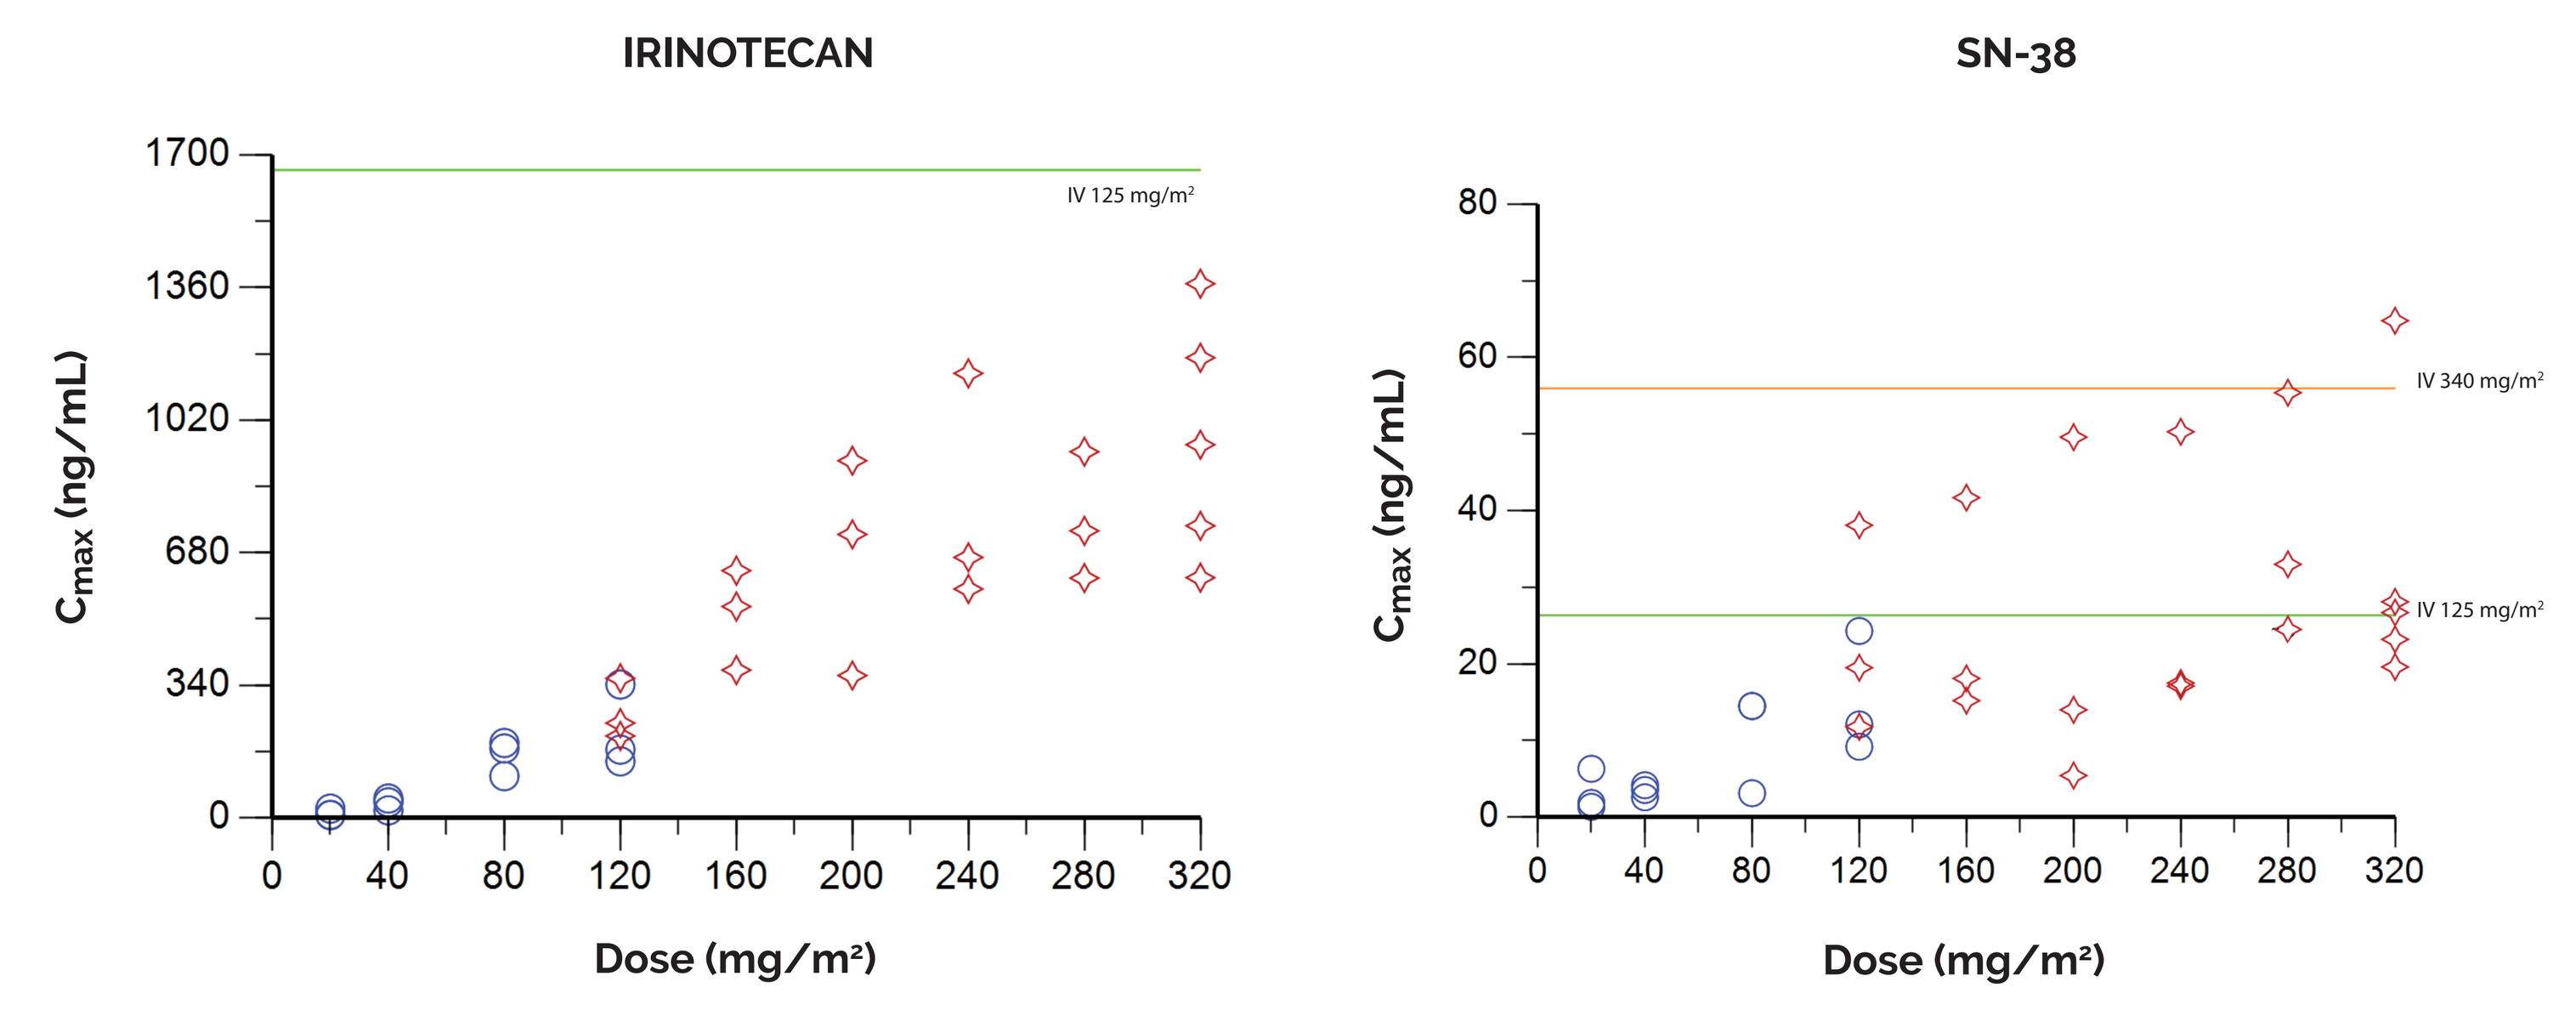
IRINOTECAN
SN-38
C_{max} (ng/mL)
Dose (mg/m²)
IV 125 mg/m²
IV 340 mg/m²
SOLUTION
TABLET

Irinotecan reference lines[a]:
IV 125 mg/m²: 1660 ng/mL
IV 340 mg/m² : 3392 ng/mL (beyond axis limits)

SN-38 reference lines[a]:
IV 125 mg/m²: 26.3 ng/mL
IV 340 mg/m²: 56.0 ng/mL

C_{max}, maximum concentration; IV, intravenous.
[a]CAMPTOSAR® (Irinotecan) Injection, intravenous infusion, Pfizer Inc., 02/2019

Plasma Pharmacokinetics: AUC_{24h} Comparison

- Half of the subjects at dose levels 200 mg/m² and above have SN38 exposures (AUC_{24h}) comparable to those in the Camptosar® label.

Figure 5.

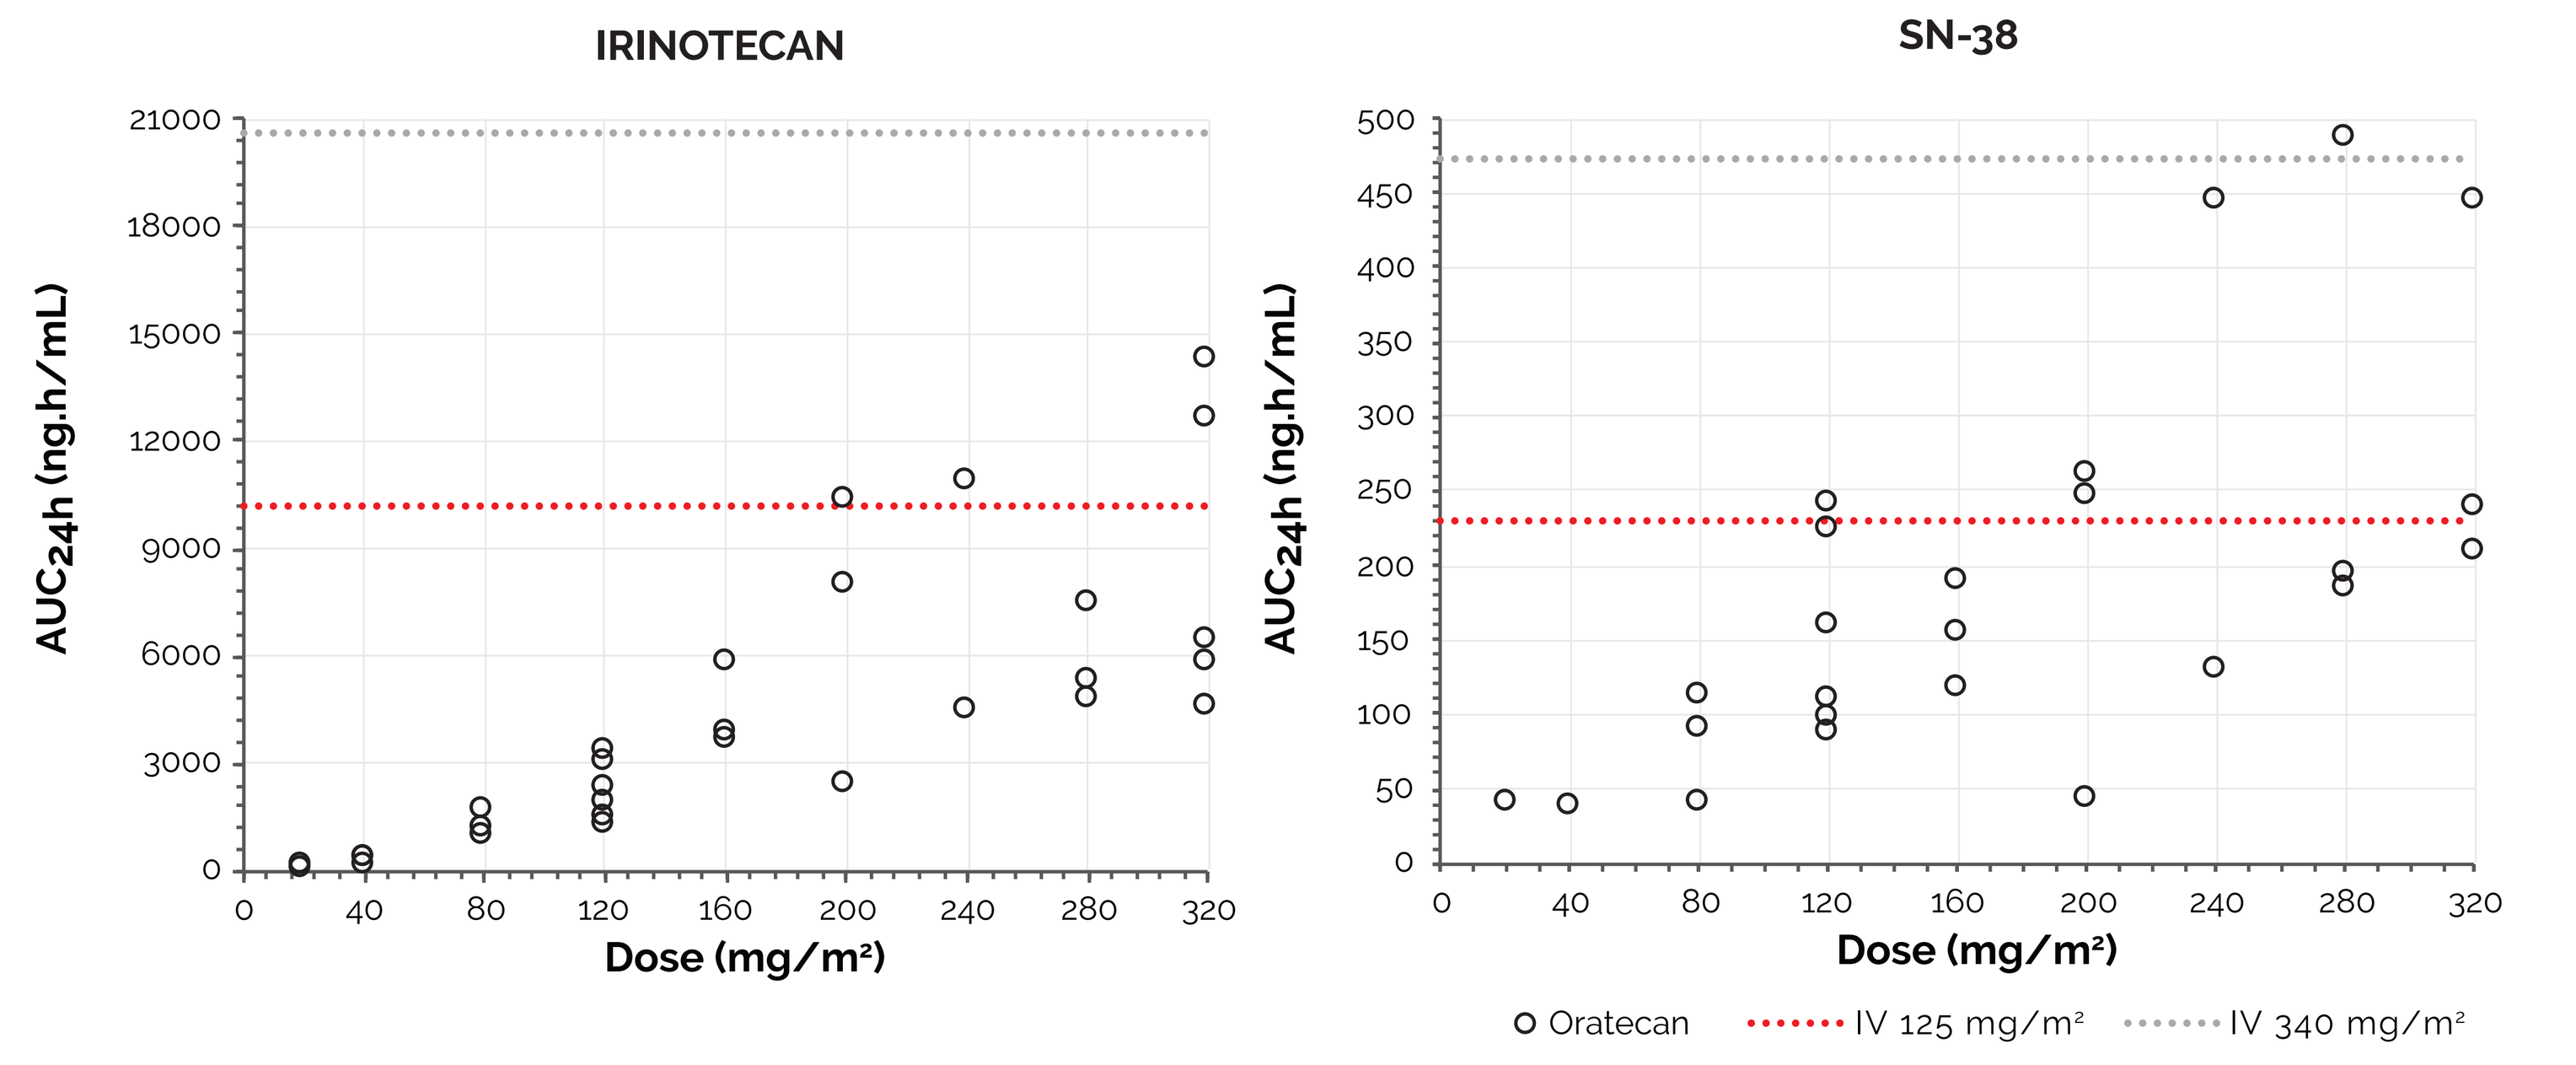
IRINOTECAN
SN-38
AUC_{24h} (ng.h/mL)
Dose (mg/m²)
Oratecan
IV 125 mg/m²
IV 340 mg/m²

Irinotecan AUC_{24h} reference lines[a]:
IV 125 mg/m²: 10,200 ng*h/mL; IV 340 mg/m² : 20,604 ng*h/mL (beyond axis limits)

SN3-8 AUC_{24h} reference lines[a]:
IV 125 mg/m²: 229 ng*h/mL; IV 340 mg/m² : 474 ng*h/mL

AUC_{24h}, 24 hour area under the concentration-time curve; IV, Intravenous.
[a]CAMPTOSAR® (Irinotecan) Injection, intravenous infusion, Pfizer Inc., 02/2019

Pharmacokinetic Summary and Comparison with Intravenous Irinotecan

Table 1. Mean (SD) pharmacokinetic parameters at the 3 highest doses tested

Dose (mg/m²) n = 3	Irinotecan				SN-38			
	t_{max} (h)	C_{max} (ng/mL)	AUC_{24h} (ng·h/mL)	$t_{½}$ (h)	t_{max} (h)	C_{max} (ng/mL)	AUC_{24h} (ng·h/mL)	$t_{½}$ (h)
240	3.0	798 (299)	6960 (3767)	11.1	3.0	28.3 (19.1)	264 (175)	20.0
280	2.0	763 (164)	6040 (1458)	12.2	1.5	37.6 (16.0)	293 (176)	18.5
320	3.0	1055 (392)	11,297 (4128)	18.7	2.0	38.5 (22.7)	300 (127)	20.4

AUC_{24}, area under the concentration-time curve at 24 hours; C_{max}, maximum concentration; $t_{1/2}$, half-life.

- When combined, C_{max} and AUC_{24h} (95% CI: 13-6204 ng·h/mL, n = 9) of SN-38 at the highest tested doses overlap the exposure range (AUC_{24h} 95% CI: 149-1400 ng·h/mL, adjusted for 3-week dosing interval) of 125 mg/m² x 2 of intravenous administration.
- Metabolic ratio was dose dependent: median (range).

Efficacy

- The best response was stable disease in 9/21 evaluable patients.

Safety

- Treatment-related Grade 3-4 adverse events were experienced by 12 (40%) patients. The most common adverse events were nausea 7 (23%), vomiting 6 (20%), and abdominal pain 3 (10%).
- Treatment-related serious adverse events were experienced by 6 (20%) patients (nausea or vomiting in 4 patients).
- Dose-limiting toxicities occurred in 2 patients at the 320 mg/m² dose level (neutropenia and *Clostridium difficile* diarrhea) and additional patients are being enrolled at the 280 mg/m² dose level to define the maximum tolerated dose.
- Acute cholinergic diarrhea has not been observed.

CONCLUSIONS

- Oral administration of encequidar followed by irinotecan tablets results in pharmacologically active concentrations of SN-38.
- Confirmation of the maximum tolerated dose when dosed on a 21-day cycle is ongoing.
- Phase 2 studies are being planned.

Reference

1. Data on file

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

Presented at 2019 ASCO Annual Meeting • May 31 - June 4, 2019 • Chicago, IL

Copyright © 2019 All rights reserved.