3090

Design, Engineering, and Characterization of a Novel Long-Acting (Pegylated) Single Isomer Human Arginase for Arginine-Depriving Anti-Cancer Treatment

Kuo-Ming Yu[1,*], Tammy Pui-Shi Pang[2], Murray Cutler[1], Johnson Yiu-Nam Lau[1,3], Thomas Wai-Hung Lo[3], Thomas Yun-Chung Leung[3]

[1]Athenex, Inc, Conventus Building, 1001 Main Street, Suite 600, Buffalo, New York, USA; [2]Avalon Polytom (HK) Ltd, Unit 608-613, IC Development Centre, No. 6 Science Park West Avenue, Shatin, Hong Kong; [3] State Key Laboratory of Chemical Biology and Drug Discovery, Department of Applied Biology and Chemical Technology and Lo Ka Chung Research Centre for Natural Anti-Cancer Drug Development, The Hong Kong Polytechnic University, Hung Hom, Kowloon, Hong Kong SAR, China;

*Correspondence to: Kuo-Ming Yu; e-mail: kuomingyu@athenex.com.

BACKGROUND

- Arginine deprivation therapy is an effective strategy to treat arginine-auxotrophic cancers with deficient expression of argininosuccinate synthetase, argininosuccinate lyase, or ornithine transcarbamylase[1-3]
- Arginase is one of the arginine-depriving enzymes and has been shown to inhibit tumor growth.[4] However, with a short half-life (< 30 min), native arginase is unable to substantially and durably deplete blood arginine in humans, rendering it inapplicable to cancer therapy
- To improve biological stability and increase the half-life for therapeutic applications, recombinant human arginase was conjugated with a linear or branched form of polyethylene glycol (PEG)[5]
- Pharmacokinetic (PK) and pharmacodynamic (PD) studies were done to select a candidate (PT01) for further evaluation. Additional in vitro and in vivo preclinical studies were done for further characterization of a new therapeutic class

METHODS

PEGylation of Recombinant Human Arginase

- Human arginase has 3 cysteines (at positions 45, 168, and 303). Two cysteines (168 and 303) were mutated to serines, leaving cysteine at 45 to create a single isoform of pegylated human arginase
- Linear or branched PEGs with 20 kDA or 40 kDa (**Figure 1**) were covalently attached to Cys45 of recombinant human arginase to yield arginase conjugated with linear-PEG [A20CL; PT01; Pegtomarginase (pegylated modified human arginase)] or branched-PEG (A20CY, A40CY)

Figure 1. The linear and branched forms of polyethylene glycol (PEG) used to pegylate recombinant human arginase

Supplier & Catalog No.	Chemical Structure
Methoxy PEG maleimide (20 kDa) (Sinopeg #06020101912)	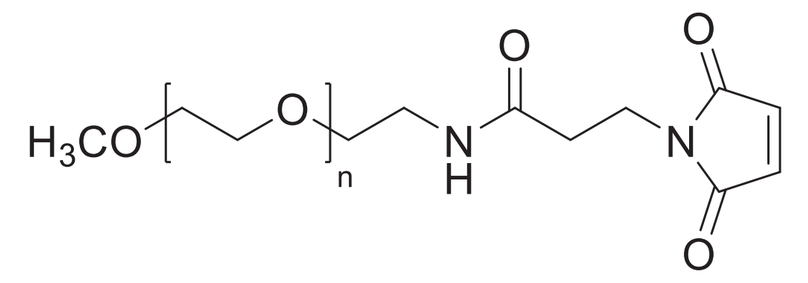
Y-shaped PEG maleimide (20 kDa) (Sinopeg #06020501954)	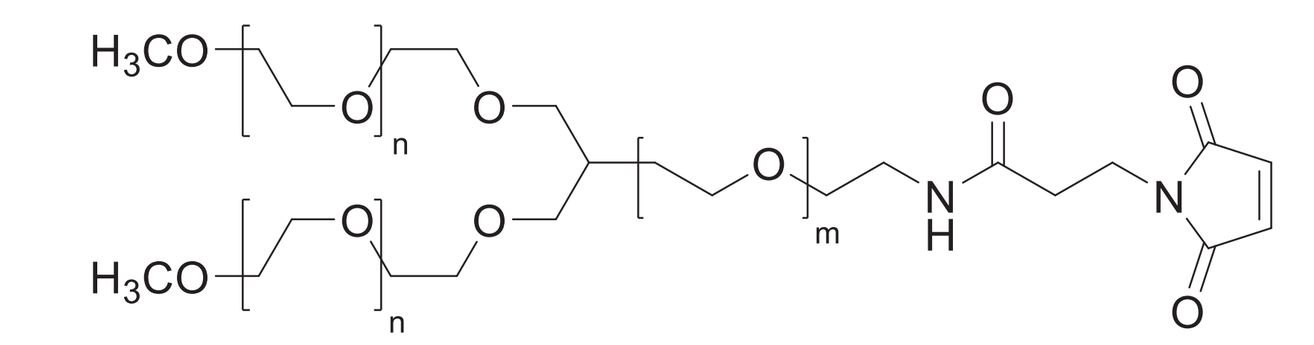
Y-shaped PEG maleimide (40 kDa) (NOF Corp. #SUNBRIGHT® GL2-400MA)	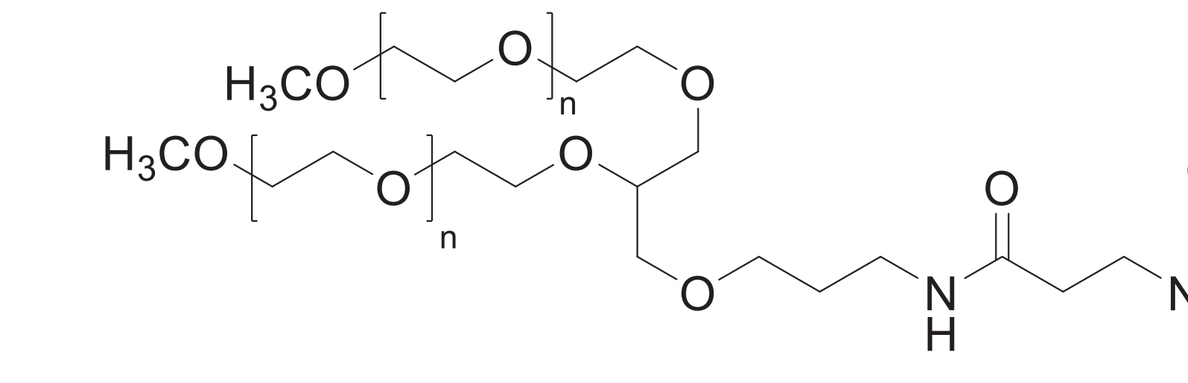

Pharmacokinetics and Pharmacodynamics of Recombinant Human Arginase Conjugated with Linear or Branched Polyethylene Glycol

- The PD properties of A20CL, A20CY, and A40CY were investigated to identify a candidate for clinical development
- The PK properties of A20CL and A20CY, as determined by enzyme-linked immunosorbent assay and catalytic activity in rats, were further investigated to confirm a candidate
- Male Sprague-Dawleys rats (n = 3) were treated with a single dose of A20CL or A20CY by intravenous (IV) injection. Body weight were taken daily, and blood samples were collected before and every 24 hours post-dose

Inhibition of Proliferation of Cancer Cell Lines by PT01

- A cell viability assay, determined with alamarBlue (Bio-Rad Laboratories, Hercules, CA, USA), was used to investigate the in vitro potency of PT01 (Pegtomarginase) to inhibit the proliferation of various cancer cell lines. The following cell lines (American Type Culture Collection (ATCC), Manassas, VA, USA) were treated with PT01 for 72 hours at 37°C at a density of 2 x 10³ cells/well in a 96-well plate:
 - HT-29 (colon cancer cell line, ATCC HTB-38, Lot 70009743)
 - MCF-7 (breast cancer cell line, ATCC HTB-22, Lot 63288596)
 - MIA PaCa-2 (pancreatic cancer cell line, ATCC CRL-1420, Lot 63143594)
 - PC-3 (prostate cancer cell line, ATCC CRL-1435, Lot 70004013)
- The percentage of inhibition was yielded as followed:

 % inhibition=[(Fluorescent signal of treated sample)/(Fluorescent signal of untreated sample)] x 100%
- The estimation of half maximal inhibitory concentration (IC_{50}) of PT01 in each cell line was calculated using an inhibitor concentration-response curve fit to the nonlinear regression model using GraphPad Prism version 7 for Mac (GraphPad Software, La Jolla, CA, USA)

In Vivo Efficacy of PT01 in Xenograft Mouse Models

- Studies were conducted to evaluate the efficacy of PT01 in xenograft mouse models commonly used in pancreatic cancer[6,7] and prostate cancer[8,9]
- Immunocompromised BALB/c nude male mice (CAnN.Cg-Foxn1nu/CrlBltw; BioLASCO Taiwan Co., Yilan, Taiwan) were inoculated with MIA PaCa-2 or PC-3 cells (5x10⁶) into the right flank of mice
- When mean tumor volume reached 200-300 mm³, mice were assigned into 4 groups (n = 8) through S-type grouping according to tumor size. Mice were treated with either vehicle (phosphate-buffered saline), gemcitabine (intraperitoneal, biweekly [BIW]) for MIA PaCa-2 or docetaxel (IV, weekly [QW]) for PC-3, or PT01. Treatments were administrated for 56 days, and the study was finished on day 63.
- Tumor size and body weight of mice were monitored and recorded twice per week
- The antitumor activities of treatments were represented as percentages of tumor growth inhibition (TGI)

 [1 – (Final tumor volume – Initial tumor volume for treated group) / (Final tumor volume – Initial tumor volume for vehicle group)] × 100
- All procedures were performed according to the Guide for the Care and Use of Laboratory Animals (National Research Council of the National Academies, Washington, DC, USA) and approved by the Institutional Animal Care and Use Committee (IACUC) of ITRI (#ITRI-IACUC-2018-040)

RESULTS

Pharmacodynamics of Recombinant Human Arginase Conjugated With Linear or Branched Polyethylene Glycol

- The potency of arginine depletion with A20CL, A20CY, and A40CY was comparable during initial 5 days post-administration (**Figure 2**)
- The costly branched 40-kDa PEG did not significantly enhance the PD effect
- Conjugates with PEGs of linear and branched 20 kDa were subjected to further evaluation

Figure 2. Plasma arginine in rats relative to pre-dose level following a dose of arginase conjugated with different polyethylene glycol moieties.

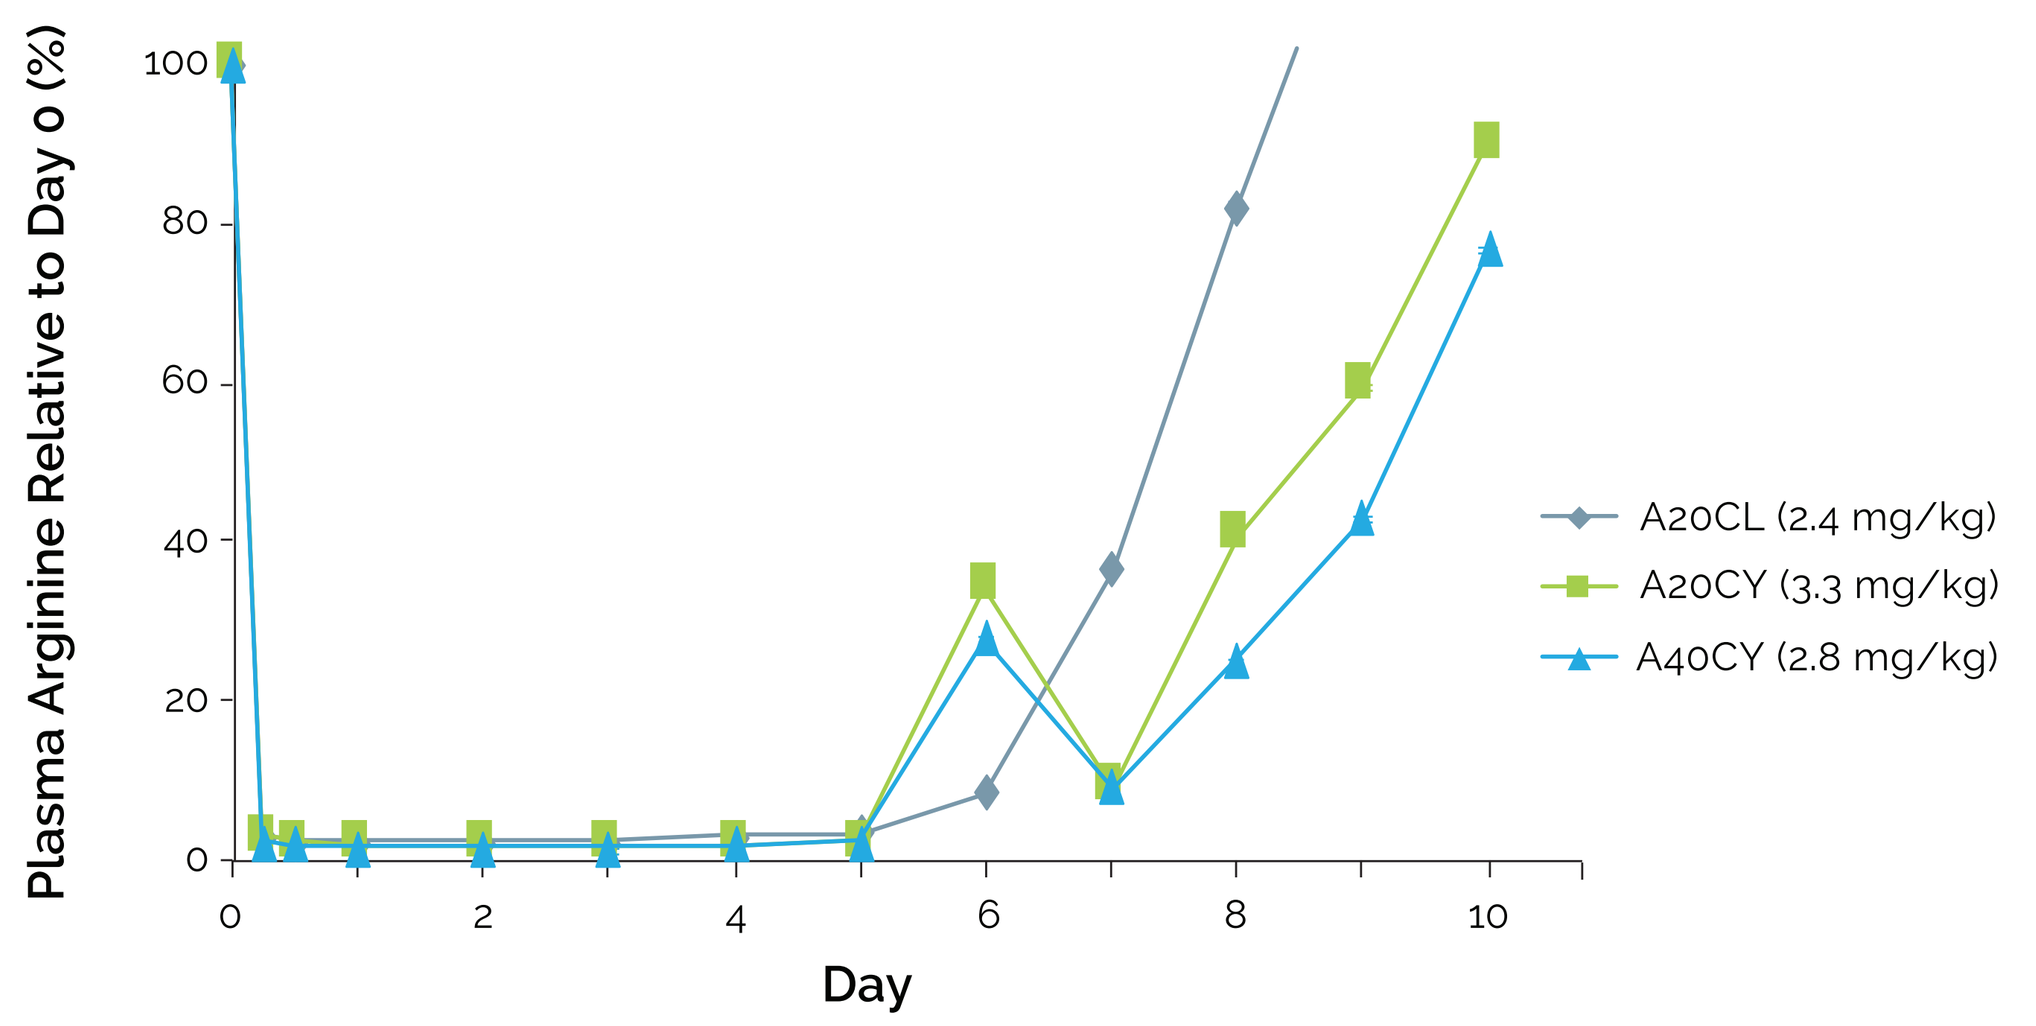
Plasma Arginine Relative to Day 0 (%)
Day
A20CL (2.4 mg/kg)
A20CY (3.3 mg/kg)
A40CY (2.8 mg/kg)

Drug Enzyme Concentration and Activity in Plasma

- Body weights of animals treated with 2 mg/kg A20CL or A20CY were comparable to those of normal Sprague-Dawley rats, suggesting that both forms of PEGylated arginase were well tolerated
- Preliminary half-lives determined by non-compartmental analysis of A20CL and A20CY were 18.8 ± 5.65 hours and 20.2 ± 10.1 hours, respectively (**Table 1**)

Table 1. Arginase activity non-compartmental analysis–derived pharmacokinetic parameters in Sprague-Dawley rats following intravenous administration

Arginase		C_{max} (mU/mL)	t_{max} (h)	t_{last} (h)	$AUC_{24h\text{-}last}$ (h*U/mL)	AUC_{last} (h*U/mL)	$AUC_{0\text{-}\infty}$ (h*U/mL)	$AUC_{\%extrap}$ (%)	$t_{1/2}$ (h)
A20CL (n = 3)	Mean	4760	24.0	104	122	317	325	2.39	18.8
	SD	420	0.00	13.9	19.4	33.9	29.7	2.72	5.65
	CV%	8.8	0.0	13.3	15.9	10.7	9.2	113.7	30.1
	Median	4630	24.0	96.0	121	308	310	1.21	19.0
A20CY (n = 3)	Mean	5240	24.0	112	130	355	364	2.50	20.2
	SD	208	0.00	13.9	21.7	1.79	7.54	2.48	10.1
	CV%	4.0	0.0	12.4	16.7	0.5	2.1	99.5	49.8
	Median	5290	24.0	120	117	355	362	2.07	17.2

AUC, area under the concentration-time curve; $AUC_{\%extrap}$, Area under the first moment of the plasma concentration-time curve extrapolated from time t to infinity as a percentage of total AUC; $AUC_{24h\text{-}last}$, area under the concentration-time curve from time 24 hours to time of last measurable concentration; AUC_{last}, area under the concentration-time curve from time 0 to time of last measurable concentration; $AUC_{0\text{-}\infty}$, area under the concentration-time curve from time 0 to infinity; CV, coefficient of variation; C_{max}, maximum concentration; SD, standard deviation; t_{last}, time of last measurable concentration; t_{max}, time of maximum concentration; $t_{1/2}$, half-life.

- The mean plasma concentrations of A20CL and A20CY 24 hours post-dose were 1010 ± 207 ng/mL and 2240 ± 446 ng/mL, respectively (**Figure 3A**). This difference is likely driven by a greater distribution of A20CL to tissues, which is reflected in a larger volume of distribution for A20CL compared with A20CY
- At 24-hours post-dose, plasma enzymatic activity in the A20CY group was slightly higher (5240 mU/mL) than that in the A20CL group (4760 mU/mL) (**Figure 3B**). The drug concentration of A20CL 24-hours post-dose was twice as high as that of A20CY; however, only an approximately 10% difference in enzymatic activity was observed
- The absence of a correlation between drug concentration and drug enzymatic activity suggests either that A20CY may not be as potent as A20CL, or that rat plasma had a higher impact on the enzymatic activity of A20CY than A20CL

Figure 3. A. Drug (PEGylated arginase) plasma concentration following a 2-mg/kg dose of A20CL or A20CY in rats. B. Drug enzyme (arginase) activity in rat plasma following a 2-mg/kg dose of A20CL or A20CY. Error bars represent standard error of the mean (SEM).

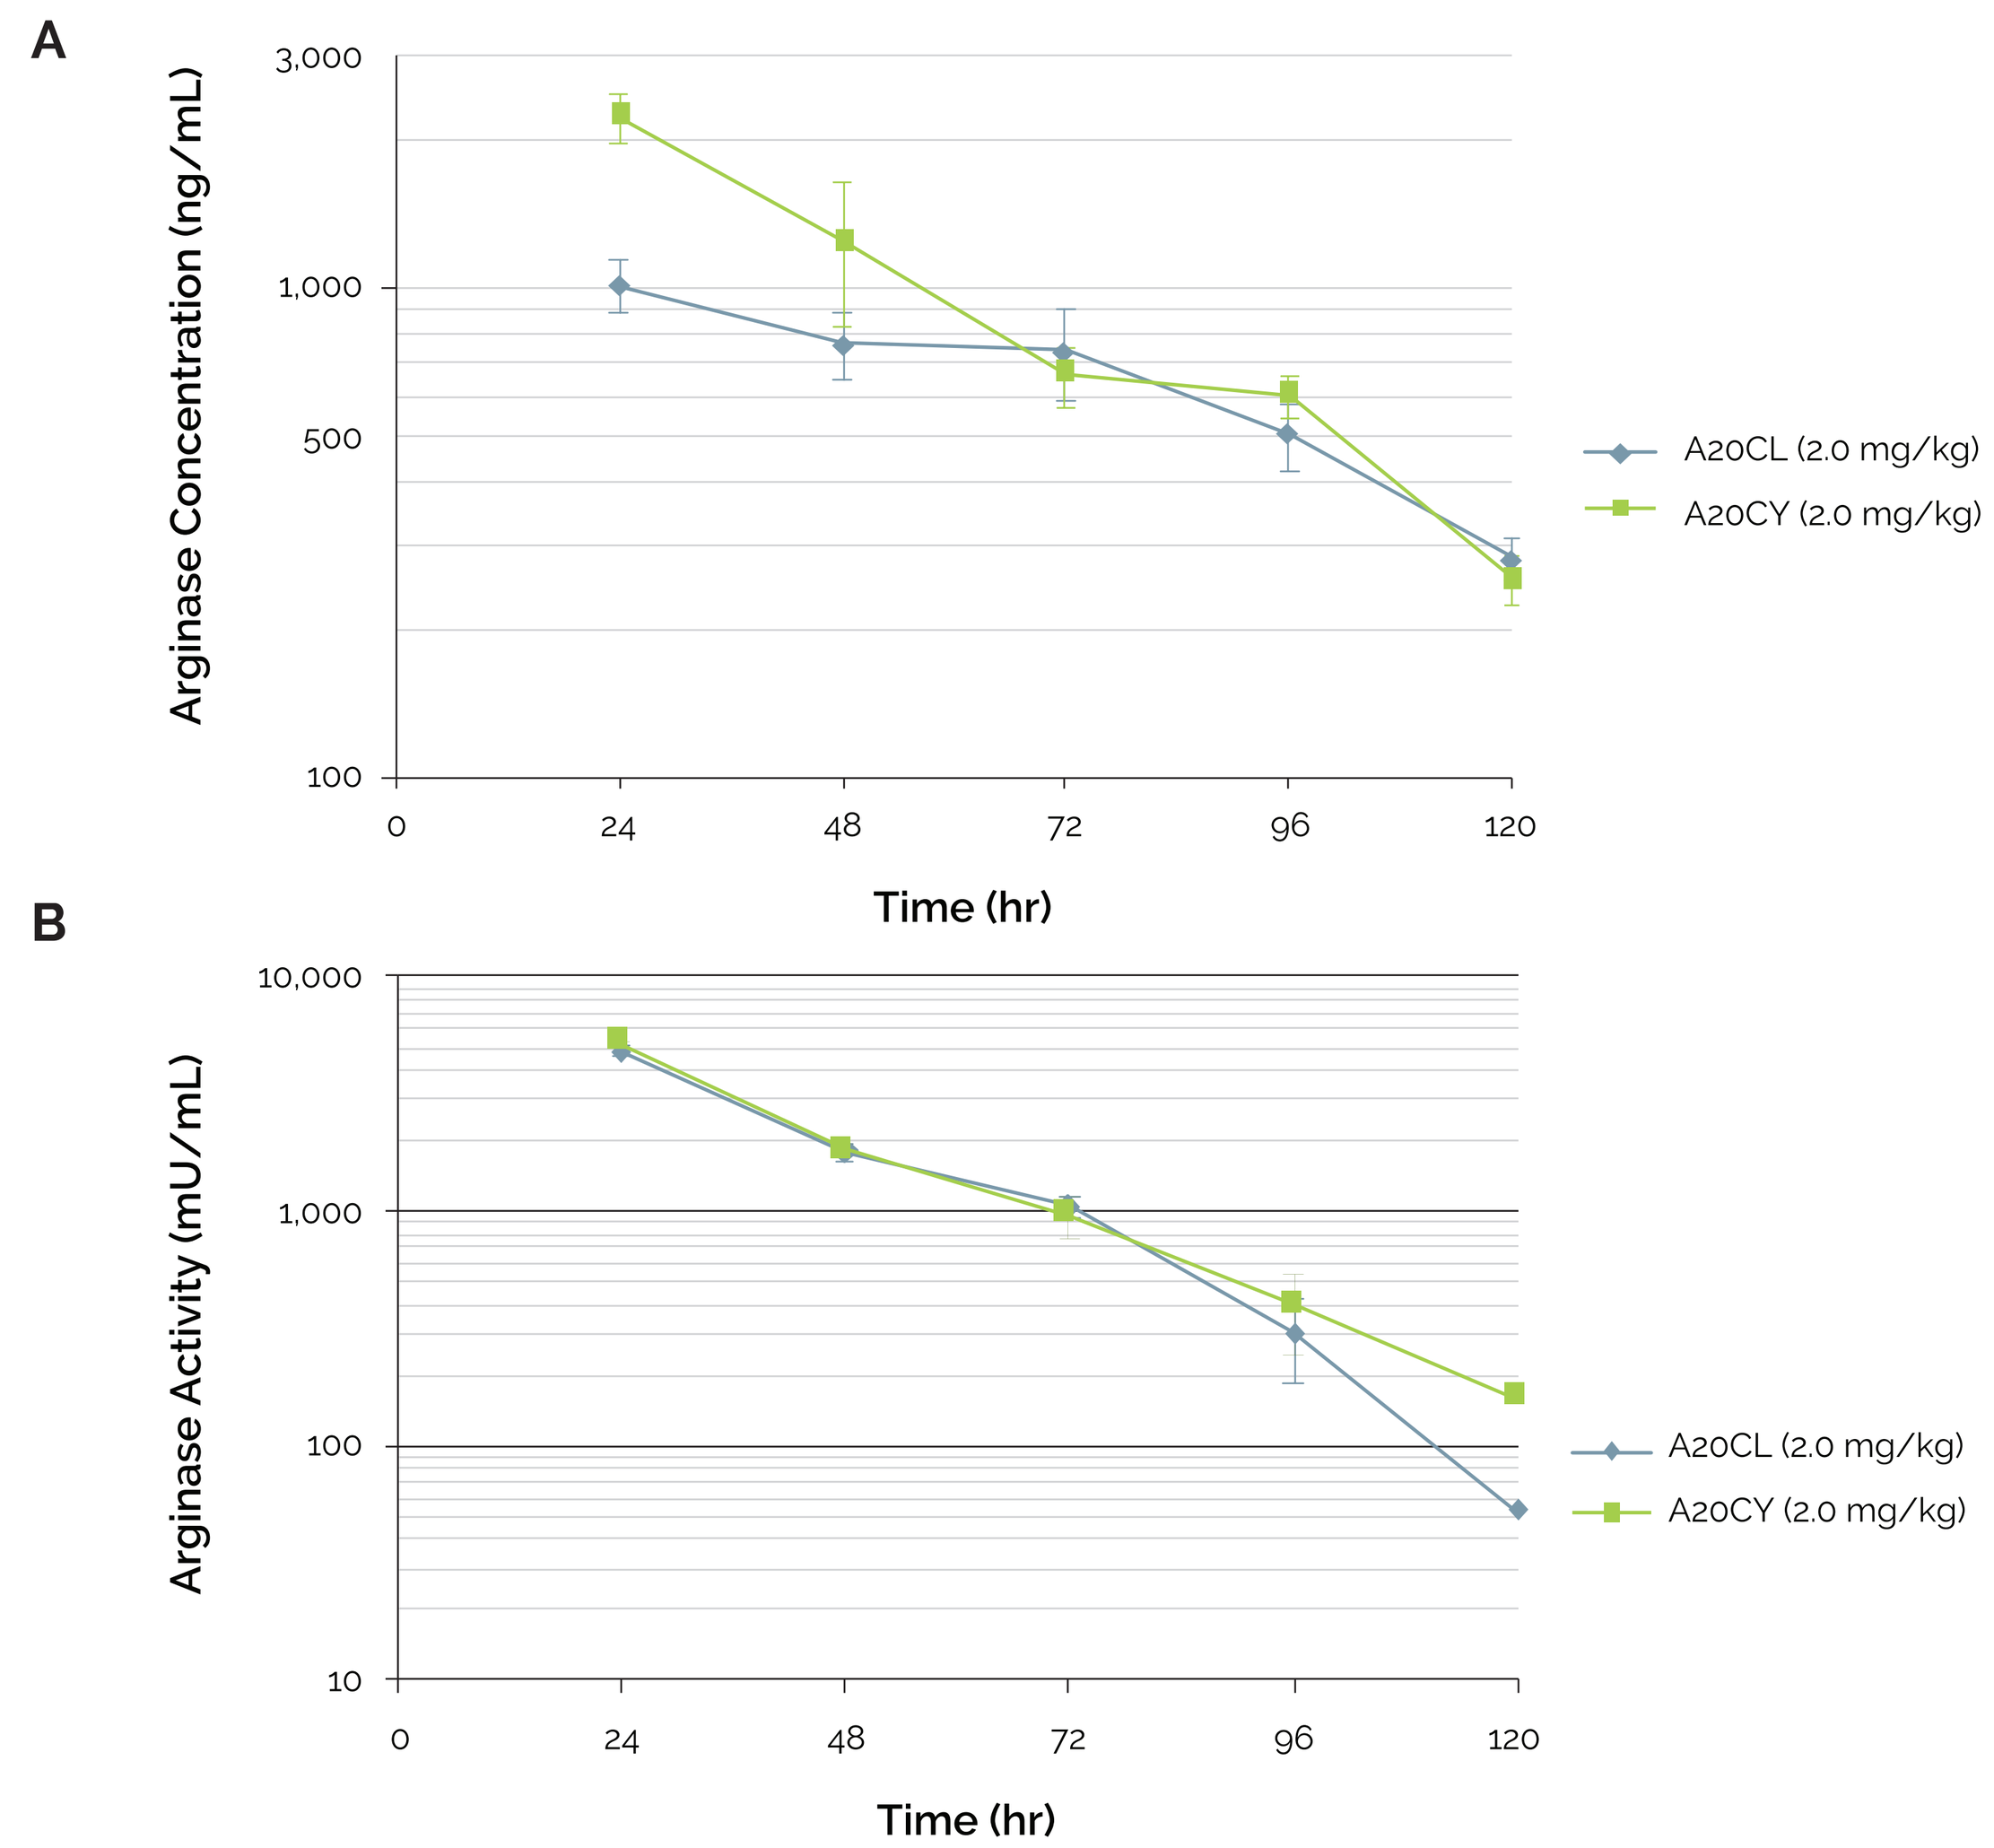
A
Arginase Concentration (ng/mL)
Time (hr)
A20CL (2.0 mg/kg)
A20CY (2.0 mg/kg)
B
Arginase Activity (mU/mL)
Time (hr)
A20CL (2.0 mg/kg)
A20CY (2.0 mg/kg)

- A dose-dependent PD effect of A20CL, as manifested on arginine depletion, was shown in **Figure 4**. Maintaining circulating arginine at low levels for at least 3 days can achieve therapeutic efficacy.[10]
- A20CL, selected as the lead candidate, showed a monoexponential decline in drug concentration and induced a rapid, near complete elimination of plasma arginine after drug administration.

Figure 4. Relative arginine plasma concentration following a dose of 0.4, 1.2, or 2 mg/kg of PT01 in rats. Error bars represent the standard error of the mean.

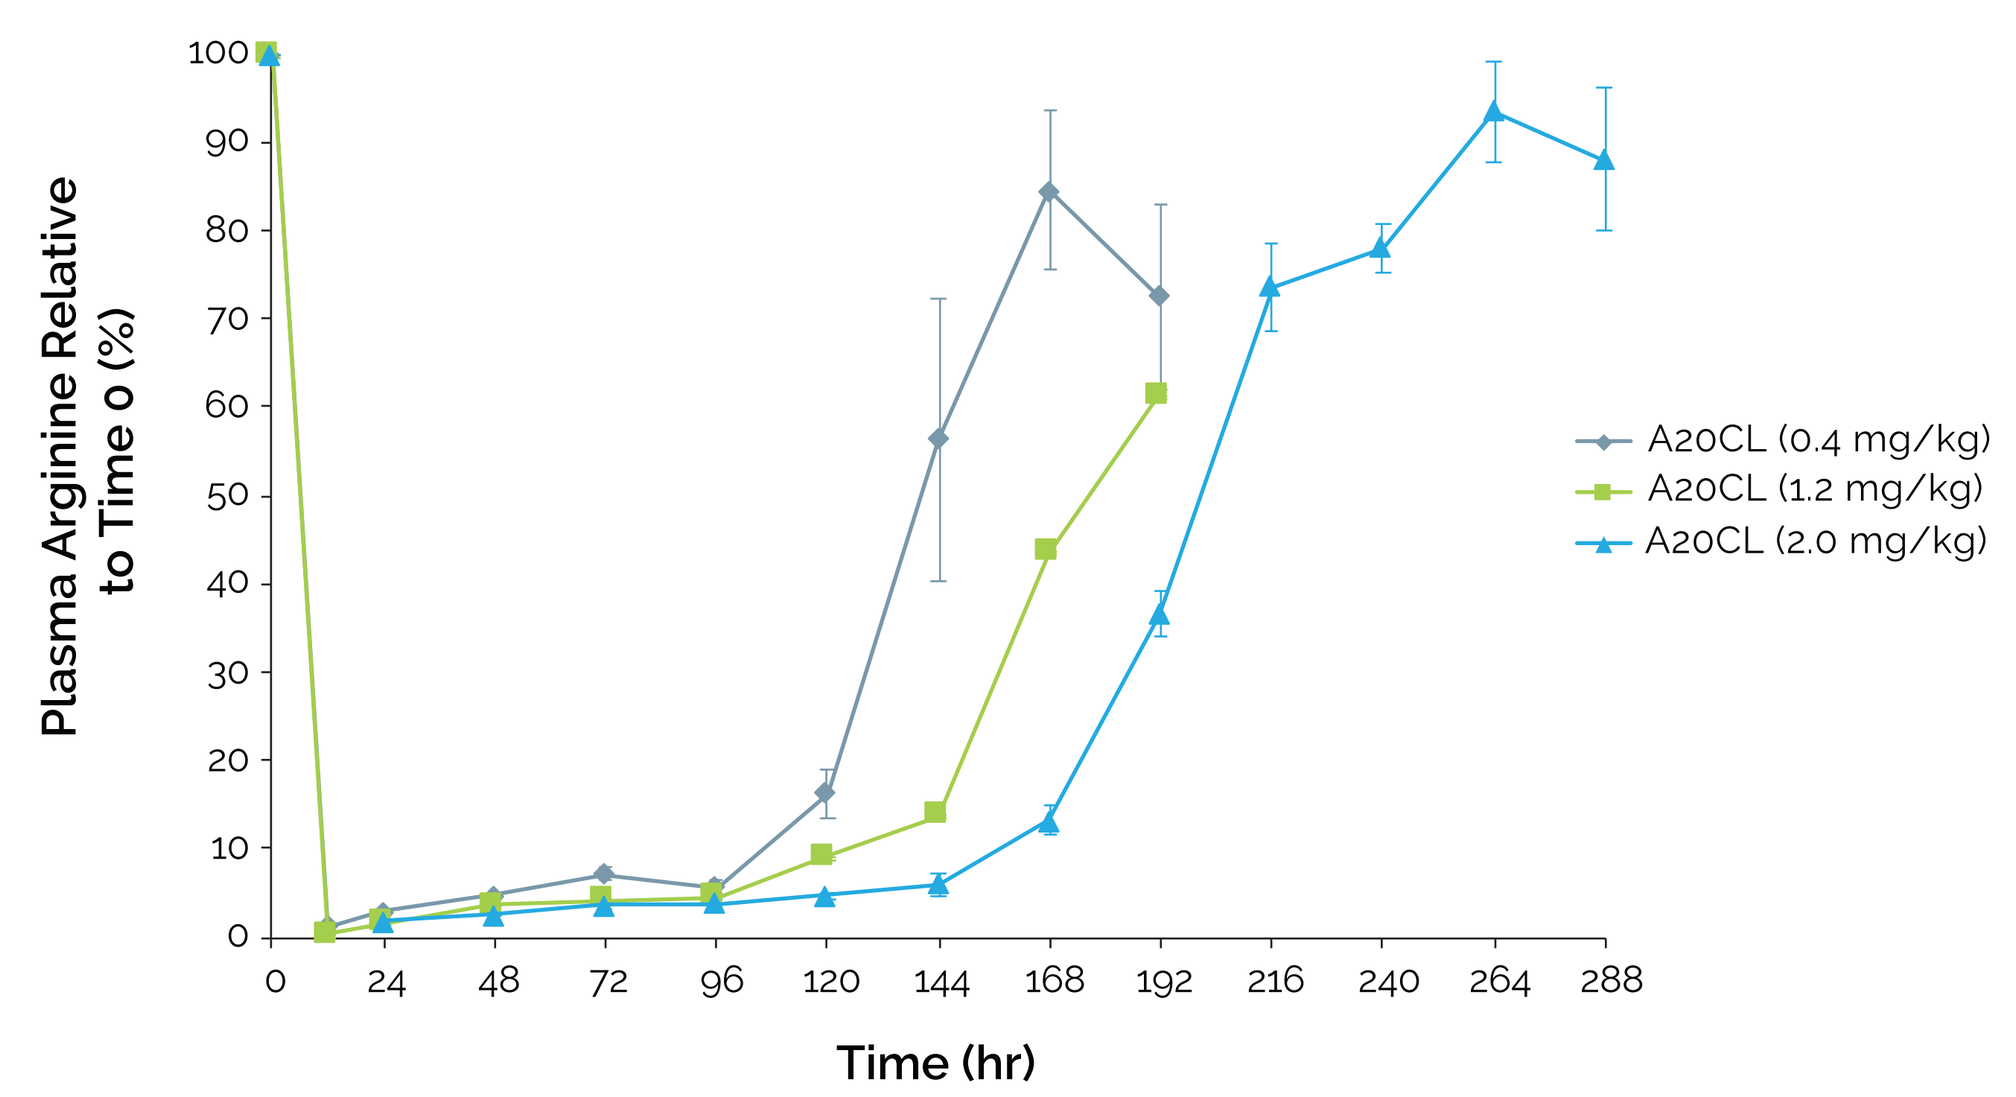
Plasma Arginine Relative to Time 0 (%)
Time (hr)
A20CL (0.4 mg/kg)
A20CL (1.2 mg/kg)
A20CL (2.0 mg/kg)

Inhibition of Proliferation of Cancer Cell Lines by PT01

- PT01 showed very potent cytotoxicity against various cancer cell lines of breast, prostate and pancreas in origins, with IC50 at sub-nM levels (**Figure 5**)
- The colon cancer cell line HT-29 was not sensitive to PT01 treatment

Figure 5. The half maximal inhibitory concentration (IC_{50}) of PT01 in PC-3, MIA PaCa-2, MCF-7, and HT-29 cancer cells

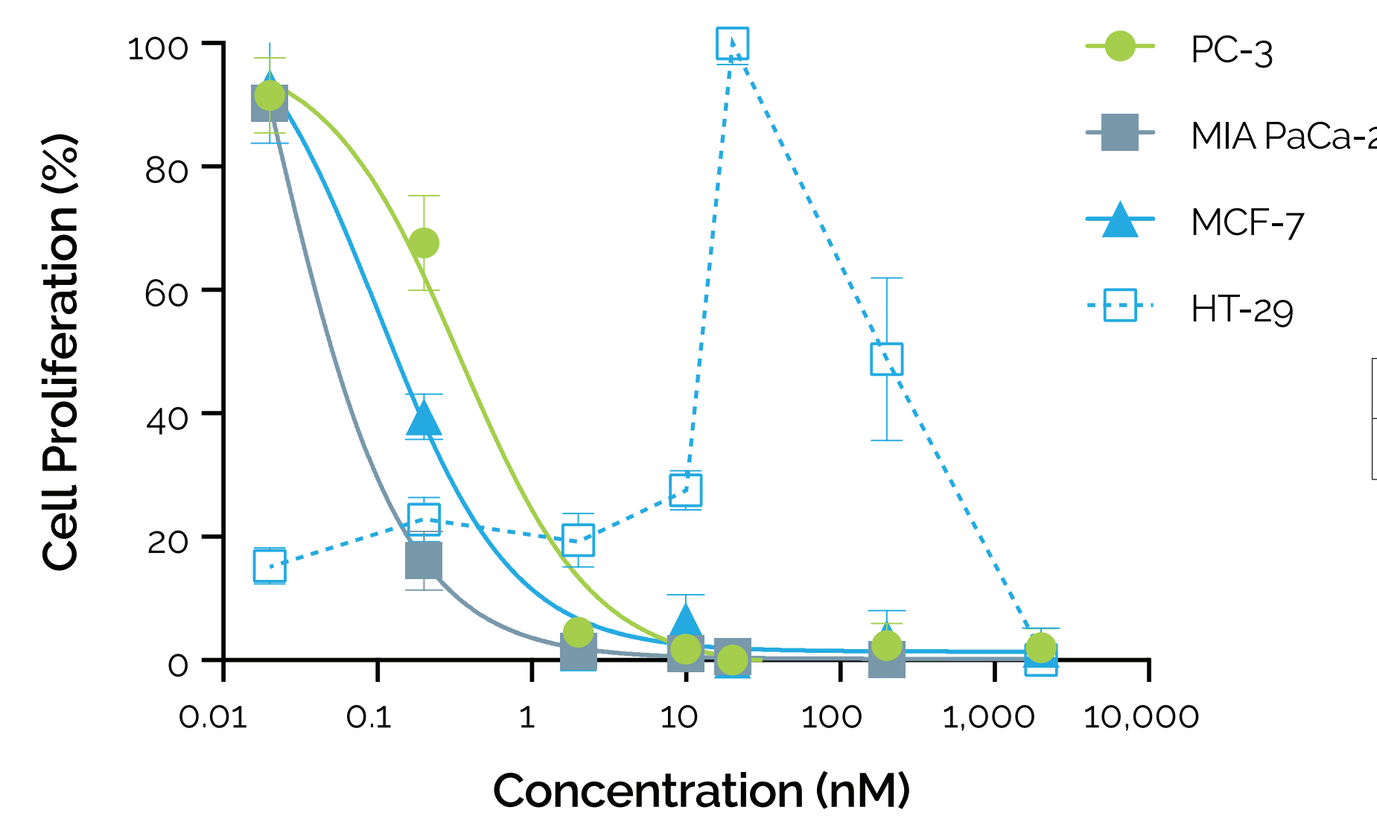
Cell Proliferation (%)
Concentration (nM)
PC-3
MIA PaCa-2
MCF-7
HT-29

	PC-3	MIA PaCa-2	MCF-7	HT-29
IC_{50} (nM)	0.32 ± 0.04	0.07 ± 0.01	0.14 ± 0.02	N/A

N/A, not applicable.

In Vivo Efficacy of PT01 in Xenograft Mouse Models

- Weekly infusion at 5 mg/kg and 10 mg/kg of PT01 induced significant TGI of 44%–67% in 2 mouse cancer models
 - Difficult-to-cure pancreatic cancer (MIA PaCa-2) (**Figure 6, Table 2**)
 - Castration-resistant prostate cancer (PC-3) (**Figure 7, Table 3**)

Figure 6. Growth of MIA PaCa-2 xenograft tumors in mice treated with vehicle, PT01 or gemcitabine (n = 8 per group, mean ± SEM).

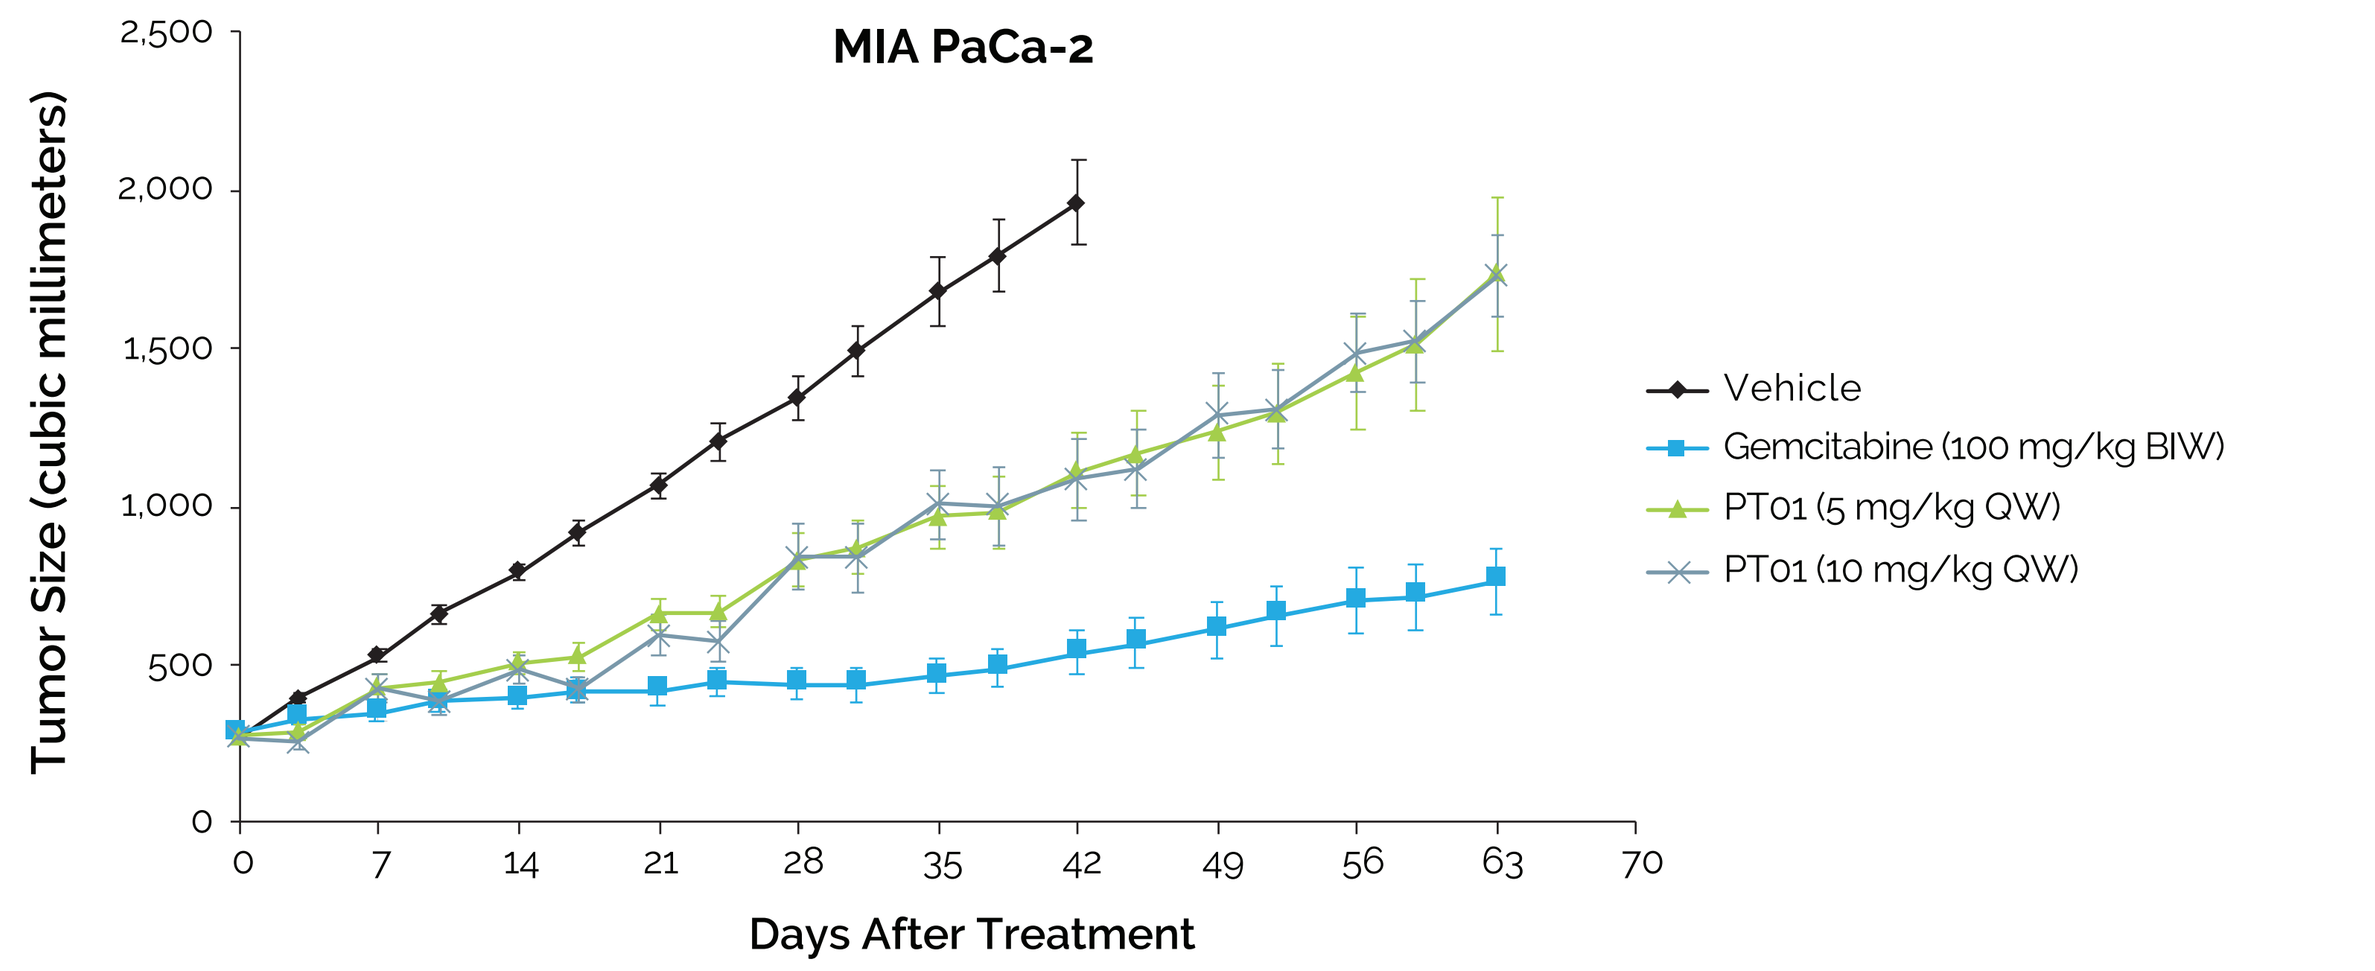
MIA PaCa-2
Tumor Size (cubic millimeters)
Days After Treatment
Vehicle
Gemcitabine (100 mg/kg BIW)
PT01 (5 mg/kg QW)
PT01 (10 mg/kg QW)

Table 2. Tumor size and tumor growth inhibition of MIA PaCa-2 xenografts following vehicle, PT01 and gemcitabine treatments

Treatment (mg/kg)	Day 42 Tumor size (mm³)	Day 42 TGI (%)	Day 63 Tumor size (mm³)
Vehicle	1960 ± 136	–	–
Gemcitabine (100 mg/kg, BIW)	537 ± 72[a]	85.0 ± 4.75	764 ± 106
PT01 (5 mg/kg, QW)	1110 ± 122[a]	50.4 ± 7.33	1740 ± 242
PT01 (10 mg/kg, QW)	1090 ± 125[a]	51.8 ± 6.56	1730 ± 130

ANOVA, analysis of variance; BIW, biweekly; IACUC, Institutional Animal Care and Use Committee; QW, weekly; TGI, tumor growth inhibition.

[a]$P < 0.0001$ compared with vehicle by one-way ANOVA; n = 8 per group. After treatments for 42 days, the mean tumor size of mice in the vehicle group reached the humane endpoints suggested by IACUC of ITRI.

Figure 7. Growth of PC-3 xenograft tumors in mice treated with vehicle, PT01 or docetaxel (n = 8 per group, mean ± SEM).

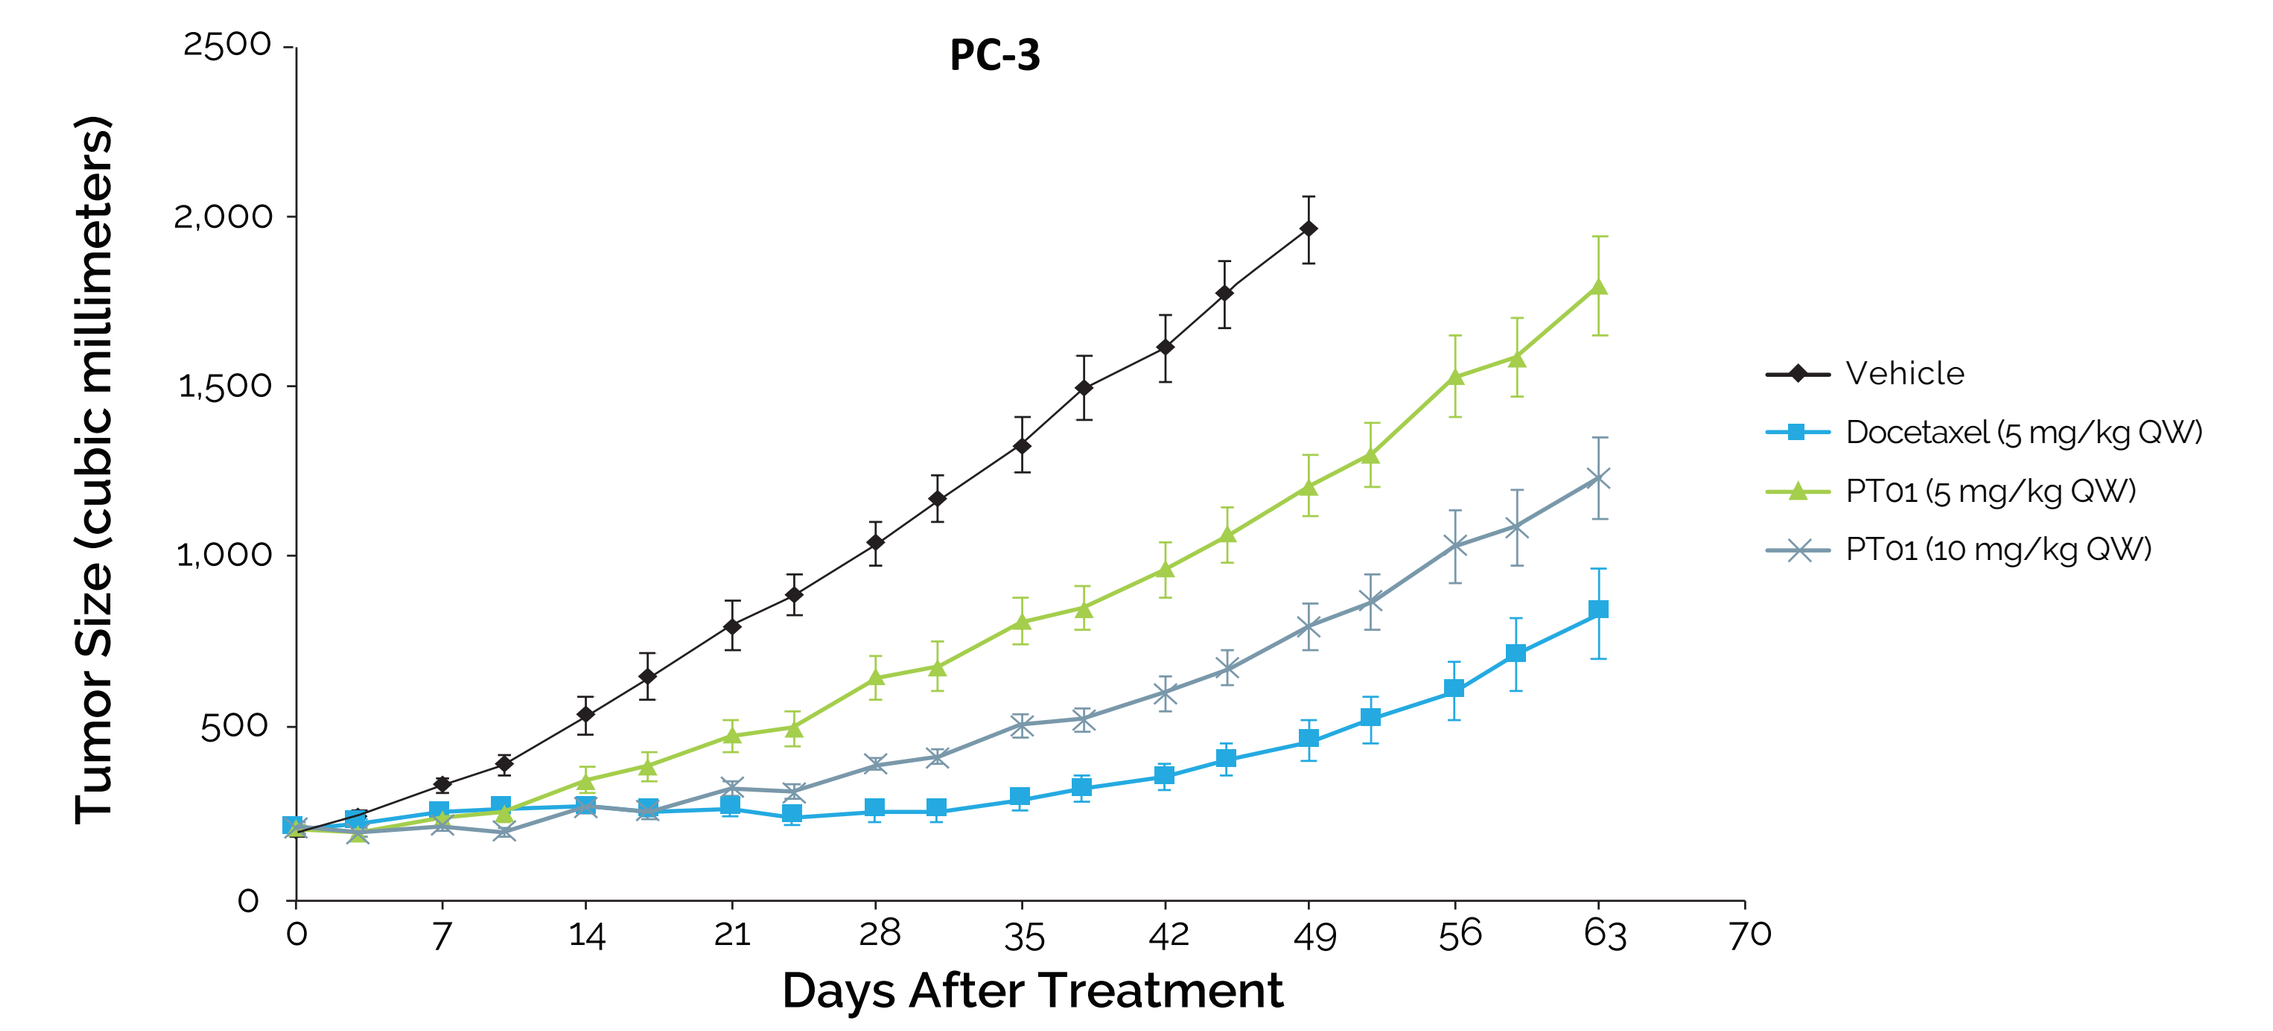
PC-3
Tumor Size (cubic millimeters)
Days After Treatment
Vehicle
Docetaxel (5 mg/kg QW)
PT01 (5 mg/kg QW)
PT01 (10 mg/kg QW)

Table 3. Tumor size and tumor growth inhibition of PC-3 xenografts following vehicle, PT01 and docetaxel treatments

Treatment (mg/kg)	Day 49 Tumor size (mm³)	Day 49 TGI (%)	Day 63 Tumor size (mm³)
Vehicle	1960 ± 96.4	–	–
Docetaxel (5 mg/kg, QW)	460 ± 57.2[a]	85.5 ± 2.70	832 ± 131
PT01 (5 mg/kg, QW)	1200 ± 88.3[a]	43.8 ± 4.74	1780 ± 146[b]
PT01 (10 mg/kg, QW)	790 ± 69.9[a]	67.1 ± 3.54	1220 ± 115

ANOVA, analysis of variance; BIW, biweekly; IACUC, Institutional Animal Care and Use Committee; QW, weekly; TGI, tumor growth inhibition.

[a]$P < 0.0001$ compared with vehicle by one-way ANOVA; n = 8 per group on day 49. After treatments for 49 days, the mean tumor size of mice in the vehicle group reached the humane endpoints suggested by IACUC of ITRI.

[b]n = 7 in PT01 (5 mg/kg) group because 1 mouse was found dead on day 56.

- PT01 treatments reduced body weight in both the MIA PaCa-2 and PC-3 xenograft mouse models 3 days after each treatment, and the effect was dose-dependent. Body weight recovered 7 days after each treatment, suggesting PT01-induced weight loss is reversible (**Figure 8**)

Figure 8. Body weight of xenograft mice following vehicle, PT01 and gemcitabine or docetaxel treatments (n = 8 per group, mean ± SEM). A. MIA PaCa-2. B. PC-3.

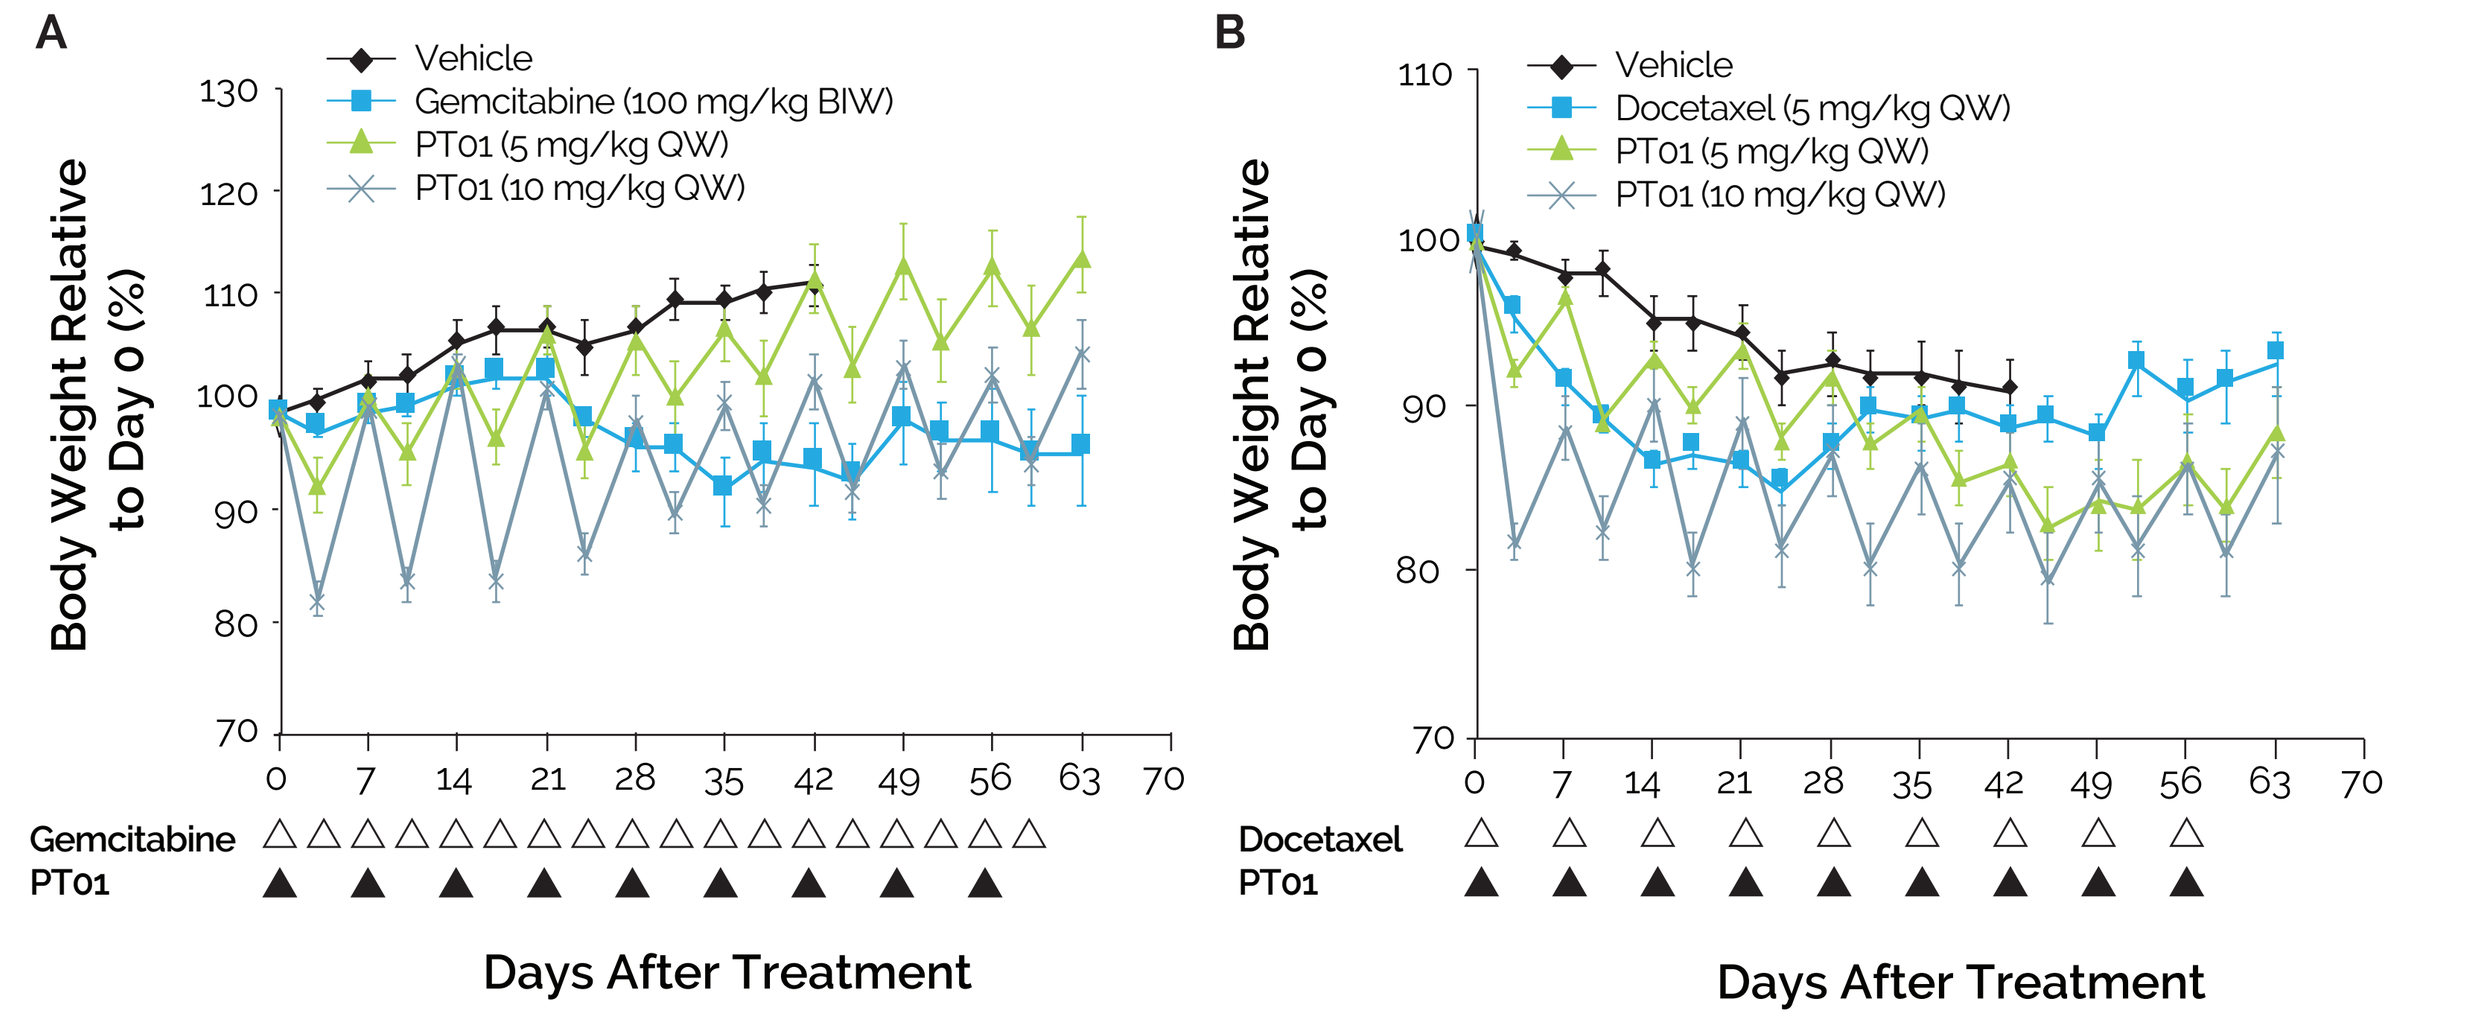
A
Body Weight Relative to Day 0 (%)
Vehicle
Gemcitabine (100 mg/kg BIW)
PT01 (5 mg/kg QW)
PT01 (10 mg/kg QW)
Gemcitabine
PT01
Days After Treatment
B
Body Weight Relative to Day 0 (%)
Vehicle
Docetaxel (5 mg/kg QW)
PT01 (5 mg/kg QW)
PT01 (10 mg/kg QW)
Docetaxel
PT01
Days After Treatment

CONCLUSIONS

- A novel single isoform of pegylated human arginase was created (PT01, Pegtomarginase) that showed excellent enzymatic activity and PK/PD profiles, maintaining circulating arginine at low levels to potentially achieve therapeutic efficacy
- PT01 exhibited potent cytotoxicity at sub-nM levels against various cancer cell lines of breast, prostate, and pancreas in origins
- PT01 demonstrated good TGI activity with tolerable toxicity in mouse xenograft models of difficult-to-cure pancreatic cancer and castration-resistant prostate cancer
- Results of the preclinical studies support further clinical development of PT01

References

1. Fung MKL, Chan GC. Drug-induced amino acid deprivation as strategy for cancer therapy. *J Hematol Oncol.* 2017;10(1):144.
2. Thongkum A, Wu C, Li YY, Wangpaichitr M, Navasumrit P, Parnlob V, et al. The Combination of Arginine Deprivation and 5-Fluorouracil Improves Therapeutic Efficacy in Argininosuccinate Synthetase Negative Hepatocellular Carcinoma. *Int J Mol Sci.* 2017;18(6). pii: E1175.
3. Riess C, Shokraie F, Classen CF, Kreikemeyer B, Fiedler T, Junghanss C, Maletzki C. Arginine-Depleting Enzymes - An Increasingly Recognized Treatment Strategy for Therapy-Refractory Malignancies. *Cell Physiol Biochem.* 2018;51(2):854-870.
4. Bach SJ, Swaine D. The effect of arginase on the retardation of tumour growth. *Br J Cancer.* 1965;19:379-386.
5. Veronese FM, Caliceti P, Schiavon O. Branched and linear poly (ethylene glycol): influence of the polymer structure on enzymological, pharmacokinetic, and immunological properties of protein conjugates. *J Bioactive & Compatible Polymers.* 1997;12(3):196-207.
6. Du Y, Shang BY, Sheng WJ, Zhang SH, Li Y, Miao QF, Zhen YS. A recombinantly tailored -defensin that displays intensive macropinocytosis-mediated uptake exerting potent efficacy against K-Ras mutant pancreatic cancer. *Oncotarget.* 2016;7(36):58418-58434.
7. Yoon DH, Shin JS, Jin DH, Hong SW, Jung KA, Kim SM, Hong YS, Kim KP, Lee JL, Suh C, Lee JS, Kim TW. The survivin suppressant YM155 potentiates chemosensitivity to gemcitabine in the human pancreatic cancer cell line MiaPaCa-2. *Anticancer Res.* 2012;32(5):1681-1688.
8. Shih TC, Liu R, Wu CT, Li X, Xiao W, Deng X, Kiss S, Wang T, Chen XJ, Carney R, Kung HJ, Duan Y, Ghosh PM, Lam KS. Targeting galectin-1 impairs castration-resistant prostate cancer progression and invasion. *Clin Cancer Res.* 2018;24(17)4319-4331.
9. Yu, XL, Jing T, Zhao H, Li PJ, Xu WH, Shang FF. Curcumin inhibits expression of inhibitor of DNA binding 1 in PC3 cells and xenografts. *Asian Pac J Cancer Prev.* 2014;15(3):1465-1470.
10. Cheng PN, Lam TL, Lam WM, Tsui SM, Cheng AW, Lo WH, Leung YC. Pegylated recombinant human arginase (rhArg-peg5,000mw) inhibits the in vitro and in vivo proliferation of human hepatocellular carcinoma through arginine depletion. *Cancer Res.* 2007;67(1):309–317.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

Presented at ASCO Annual Meeting • May 31 - June 4, 2019 • Chicago, IL

Copyright © 2019 All rights reserved.